TPS3131: A phase 1/2 study with birinapant in combination with pembrolizumab

Russell J. Schilder[1], Mark Albertella[2], James Strauss[3], Malin Sydvander[2], Santosh Nair[4], Kingsley Urakpo[2], Stefan Norin[2] and John Öhd[2]
[1]Department of Medical Oncology, Thomas Jefferson University, Philadelphia, PA, USA, [2]Medivir AB, Huddinge, Sweden, [3]Mary Crowley Cancer Research Center, Dallas. TX, USA, [4]Mid Florida Cancer Centers, Orange City, FL, USA

Study rationale

- The inhibitor of apoptosis protein (IAP) genes are amplified in a large number of tumor types including cervical, ovarian, head and neck, pancreatic and esophageal tumors
- Overexpression of IAPs has been linked to resistance to chemotherapy and radiation
- Birinapant is a bivalent SMAC mimetic active against multiple IAP family members including cIAP1, and has demonstrated clinical tolerability with robust and durable target engagement in patients with advanced cancers
- Birinapant and immune checkpoint inhibitors are synergistic in preclinical models, consistent with the reported role of cIAP1 in tumor cells and immune cells
- Based on these observations, a phase 1/2 trial with birinapant and pembrolizumab has been initiated (NCT02587962) and opened for enrollment in August 2017

Birinapant and pembrolizumab – potential synergy

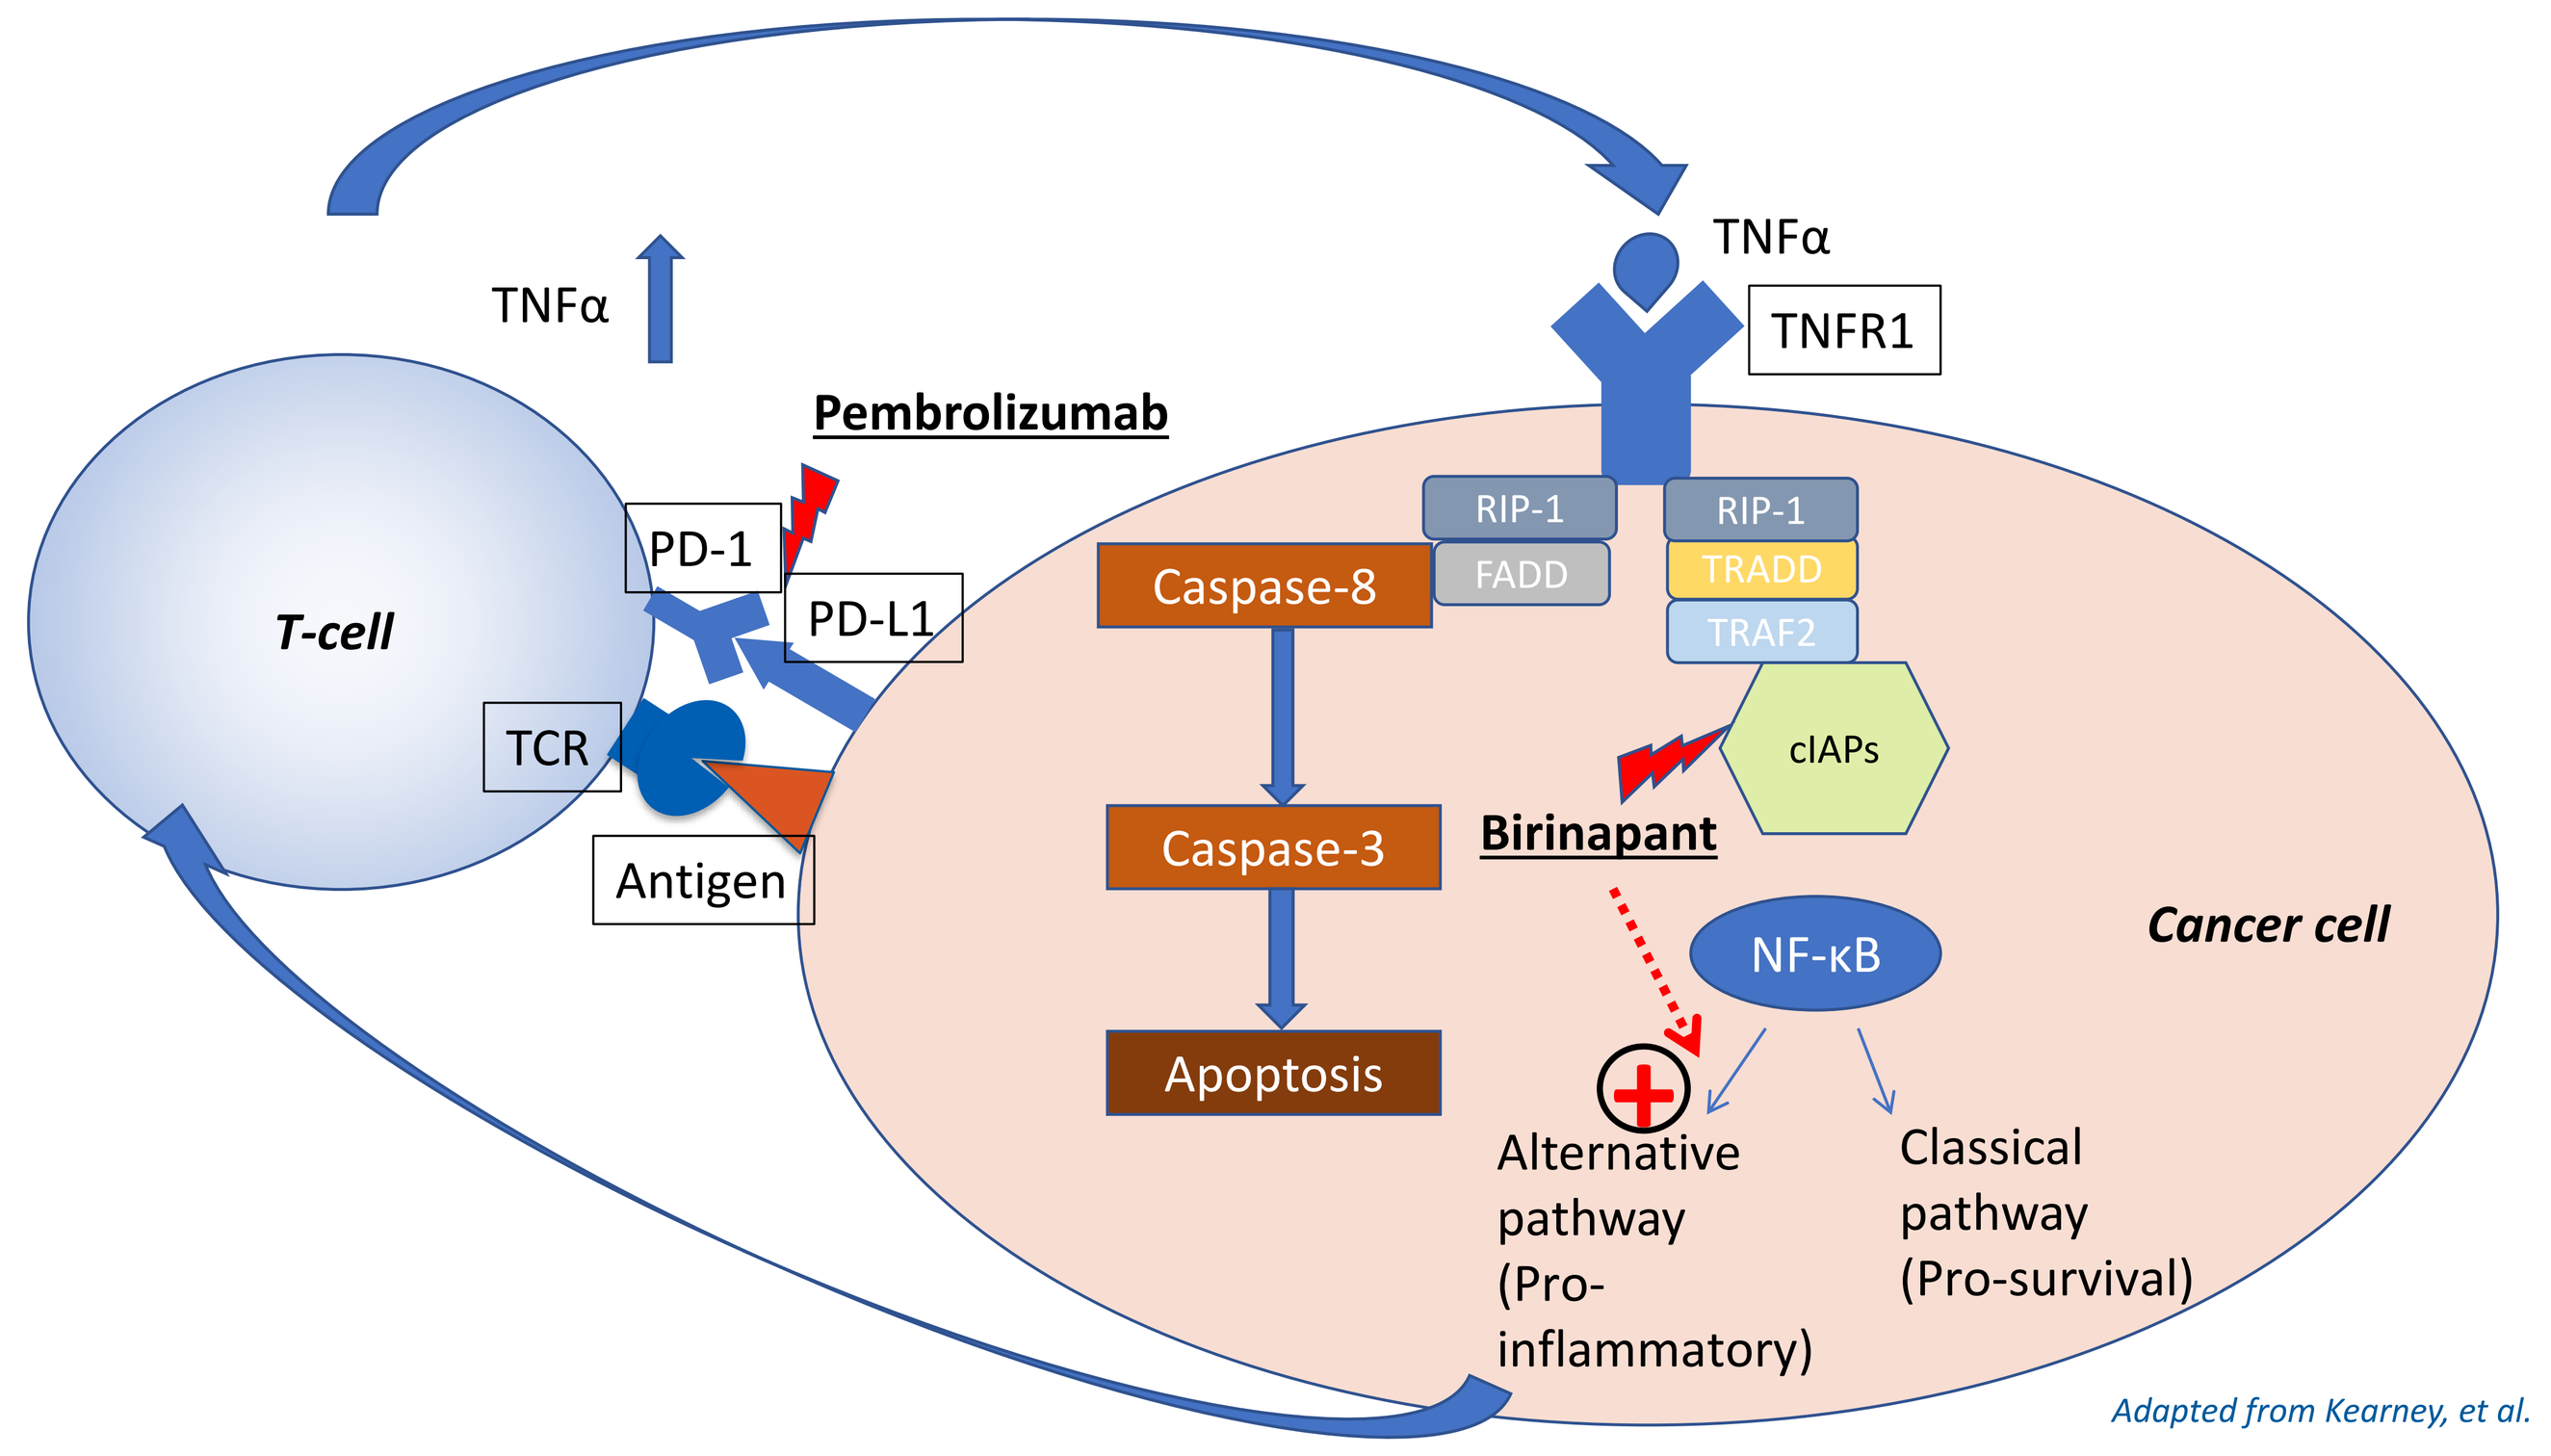
TNFα
TNFα
TNFR1
Pembrolizumab
T-cell
PD-1
PD-L1
TCR
Antigen
Caspase-8
RIP-1
FADD
RIP-1
TRADD
TRAF2
cIAPs
Caspase-3
Birinapant
Apoptosis
NF-κB
Cancer cell
Alternative pathway (Pro-inflammatory)
Classical pathway (Pro-survival)

Adapted from Kearney, et al.

- Birinapant binds to cIAP proteins and causes their auto-ubiquitination and proteasomal degradation
- Destruction of cIAP1 by birinapant switches TNFR1 signaling to induce apoptosis
- PD1 blockade increases TNFα production which is converted to pro-apoptotic signal by birinapant
- cIAPs suppress alternative NF-κB signaling which is increased by birinapant and can lead to further activation of the immune system

Study Objectives and Design – Dose Escalation

- **Primary objective:** To determine the safety and tolerability of the recommended phase 2 dose (RP2D) of birinapant in combination with pembrolizumab IV
- **Secondary objective:** To assess preliminary efficacy of the combination of pembrolizumab and birinapant in patients with relapsed or refractory cancer by effects on tumor size as measured by imaging (CT or MRI) assessed by RECIST 1.1
- Standard 3+3 design
- Birinapant doses to be evaluated: 5.6, 11, 17 and 22 mg/m^2 IV on day 1 and 8 in a 21-day cycle in addition to pembrolizumab 200 mg IV on day 1
- Dose-limiting toxicity (DLT) is defined as Grade ≥3 non-hematological toxicity, excluding isolated laboratory abnormalities except grade ≥3 ASAT, ALAT or lipase elevation for > 7 days, grade 4 neutropenia, grade ≥3 febrile neutropenia or grade ≥3 thrombocytopenia with bleeding during the first treatment cycle and considered related to either birinapant or pembrolizumab
- RP2D will be proposed by the safety review committee

Study Objectives and Design – Dose Expansion

- **Primary objectives:**
 - To determine whether the combination of pembrolizumab and birinapant has sufficient antitumor activity, as assessed by ORR against colorectal, ovarian and cervical cancer to warrant more extensive development
 - To determine the safety and tolerability of the RP2D of birinapant when given in combination with pembrolizumab to patients in the various solid tumors cohort
- **Secondary objectives:**
 - To assess the safety and tolerability of the combination of pembrolizumab and birinapant; Overall and in the defined tumor types, colorectal cancer, ovarian cancer and cervical cancer
 - To assess clinical activity of the combination of pembrolizumab and birinapant in the defined tumor types by effects on tumor response, including clinical benefit rate, time to response and duration of response, assessed by RECIST 1.1
 - To assess clinical activity of the combination of pembrolizumab and birinapant in the defined tumor types by effect on overall survival
 - To assess clinical activity of the combination of pembrolizumab and birinapant in the defined tumor types by effect on progression free survival

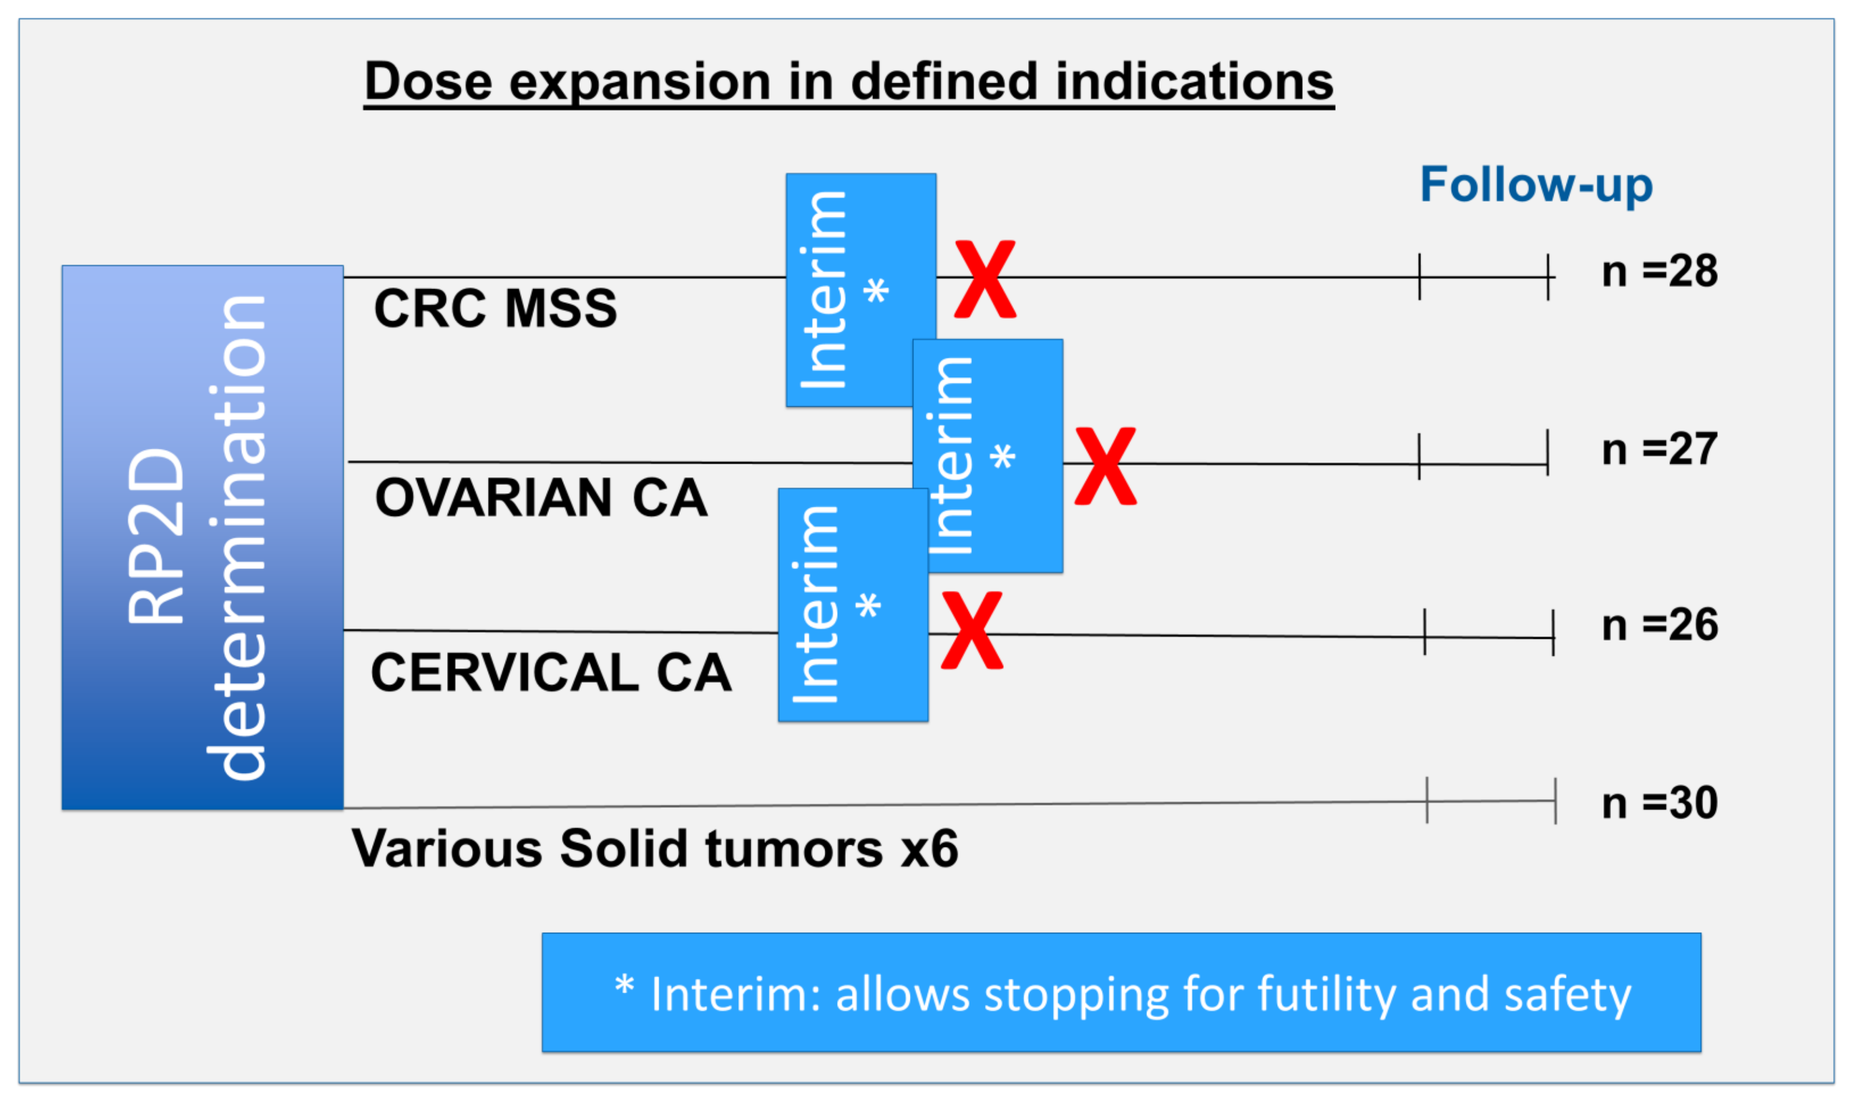
Dose expansion in defined indications
Follow-up
RP2D determination
CRC MSS
OVARIAN CA
CERVICAL CA
Various Solid tumors x6
Interim *
Interim *
Interim *
X
X
X
n =28
n =27
n =26
n =30
* Interim: allows stopping for futility and safety

- Phase 2 part is planned to include 111 patients in three separate main cohorts using a Simon's two-stage design that yields a type I error rate of 0.05 and statistical power of 0.80 for each of the three cohorts using a one-sided test based on true response rates of 20% (colorectal cancer), 25% (ovarian cancer) and 30% (cervical cancer)
- An exploratory cohort enrolling five patients each with small cell lung cancer, cholangiocarcinoma, gastroesophageal carcinoma, mesothelioma, head and neck squamous cell carcinoma (check-point inhibitor-naïve and experienced) will be included

Major inclusion criteria

Phase 1/2:

- Patients >18 years of age
- ECOG 0-1
- Amylase and lipase < Upper limit of normal
- No further suitable standard therapeutic options

Phase 1 only: Histologically confirmed diagnosis of a metastatic or unresectable solid malignancy

Phase 2 only: A histologically confirmed diagnosis of

- Microsatellite stable colorectal cancer
- Ovarian cancer
- Cervical cancer
- Head and Neck squamous cell carcinoma (HNSCC), Small cell lung cancer, Cholangiocarcinoma, Mesothelioma, Gastroesophageal carcinoma (Exploratory cohort)

Major exclusion criteria

- Prior therapy with anti-PD-1, anti-PD-L1, anti-CTLA-4 or other checkpoint inhibitors (except HNSCC, checkpoint experienced group)
- Ongoing or recent treatment with anti-TNF therapies
- Active autoimmune disease
- Active CNS metastases or carcinomatous meningitis
- History of interstitial lung disease

Pharmacokinetic and translational biomarker analyses

Exploratory objectives

The pharmacokinetics of birinapant in plasma when administered in combination with pembrolizumab will be assessed

Marker	Purpose of assessment
Blood samples	
cIAP1 protein	Evidence of target engagement Pharmacodynamic marker, well precedented in previous clinical studies.
Serum cytokines	Evidence of immune system modulation
Gene expression analysis	Evidence of immune system modulation Understand mechanism of action
Tumor samples	
PD-L1 expression	Best validated predictor of potential pembrolizumab responder
Gene expression analysis	Additional predictor of pembrolizumab sensitivity based on IFNγ signature Potential to identify other response signatures in future
cIAP1 expression	Potential predictor of birinapant sensitivity PD in tumor samples for comparison to PBMC
IAP gene copy number	Assess copy number variation of all 8 IAP genes and TNFR-associated genes
Tumor Infiltrating Lymphocytes (TILs)	Potential response marker TIL evidence of increased immune activity

Mechanism of action (PD)

Prediction of response

Example of duplex immunohistochemistry staining for cIAP1 and CD8+ T cells

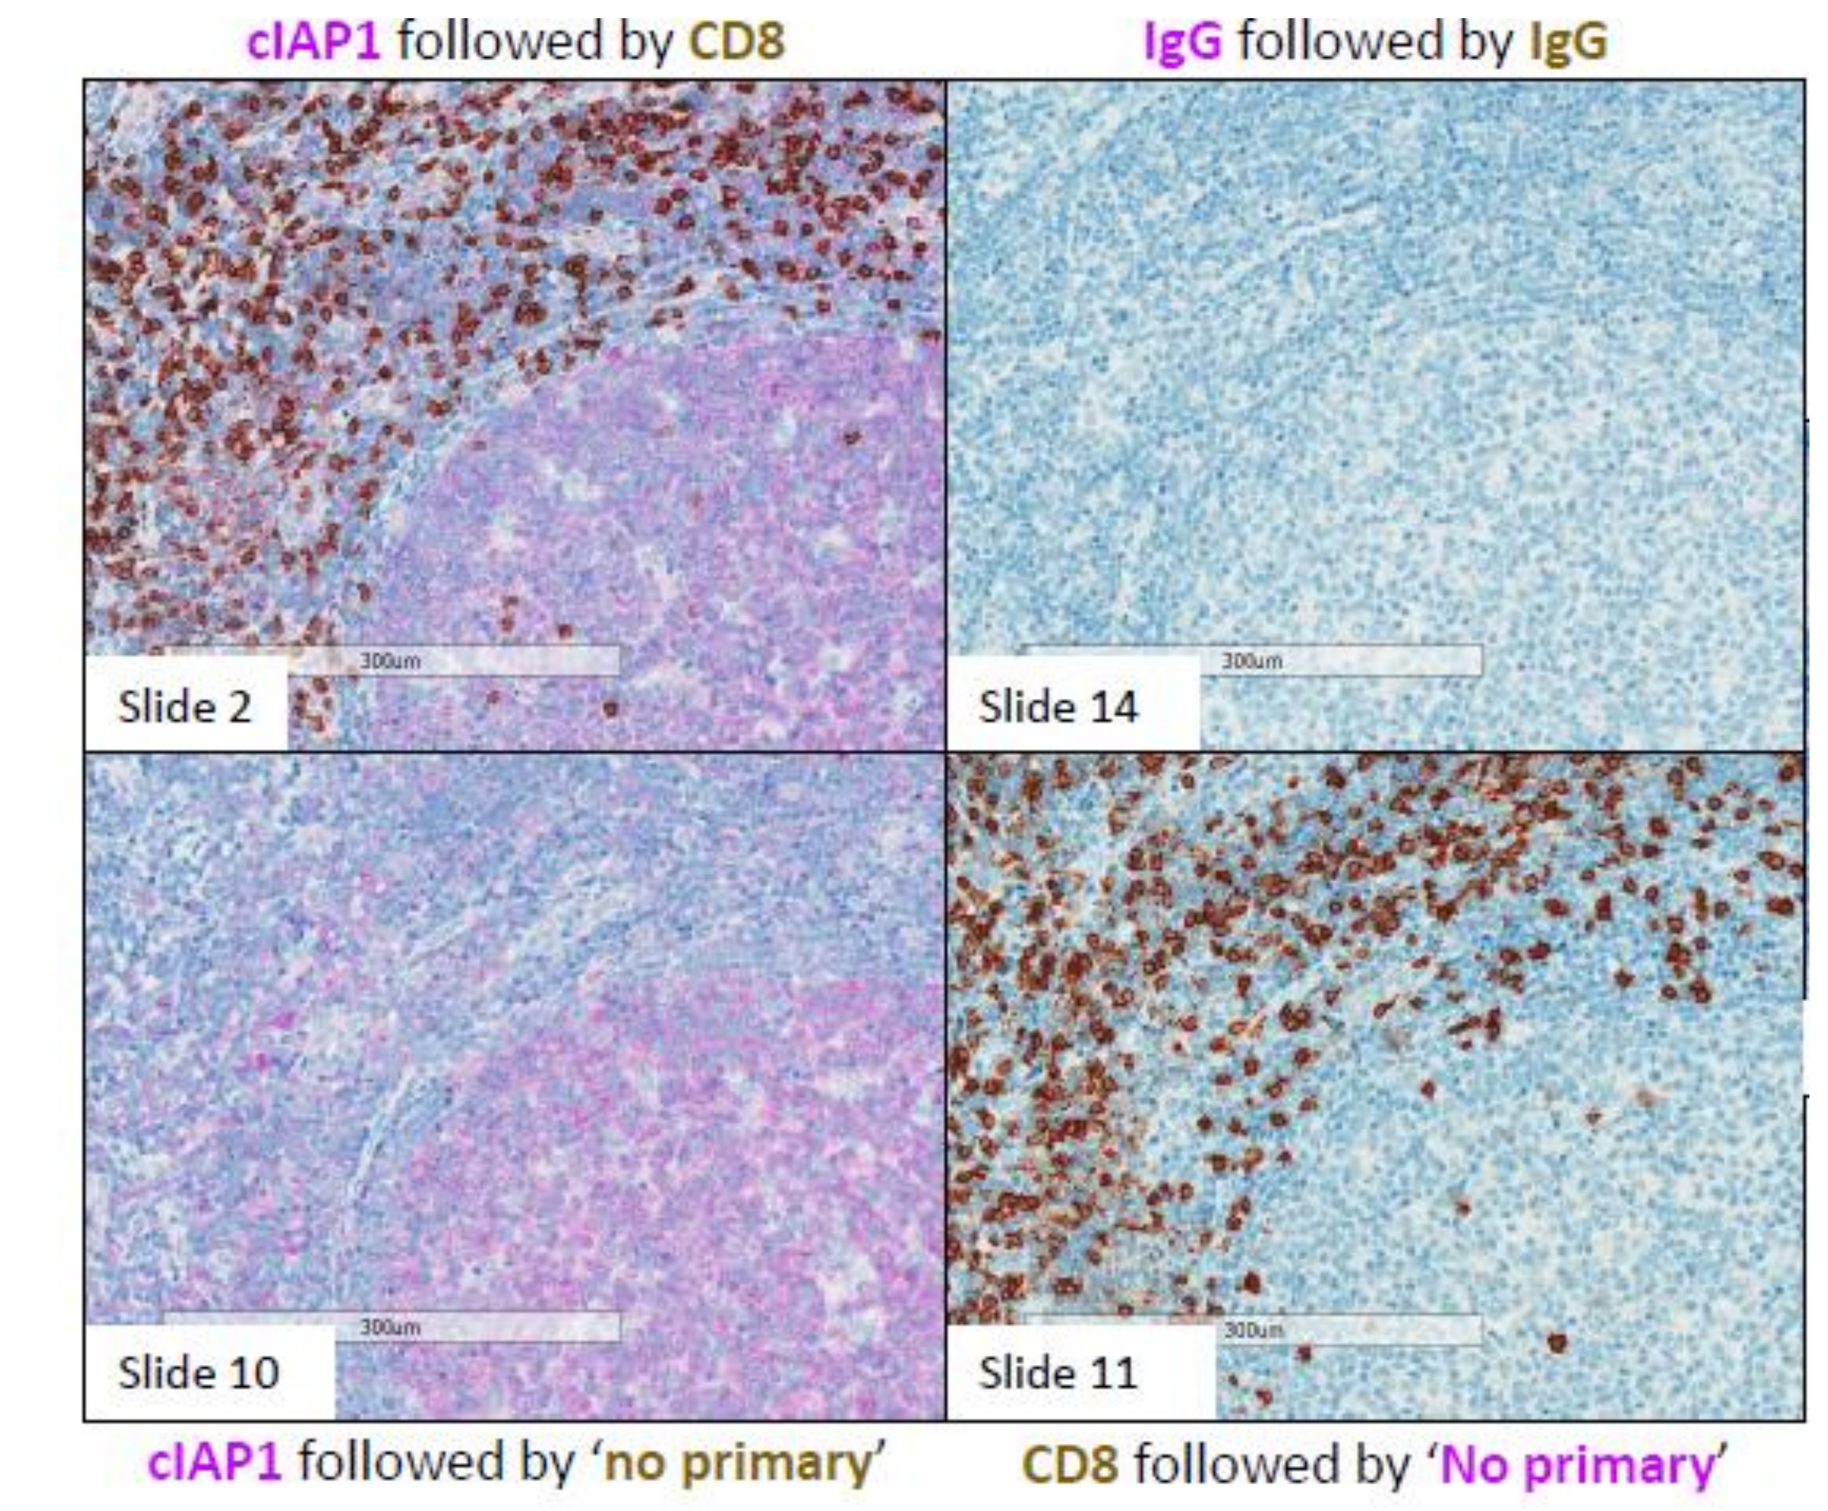
cIAP1 followed by CD8
IgG followed by IgG
Slide 2
Slide 14
Slide 10
Slide 11
cIAP1 followed by 'no primary'
CD8 followed by 'No primary'

Tonsil sample for assay validation not a study patient sample

References

1. Beug et al. Nat Commun. 2017;8:14278.
2. Condon et al. J Med Chem. 2014;57:3666-3677
3. Kearney et al. Cell Death Differ. 2017;24:1705-1716
4. Dougan et al. J Exp Med. 2010:2195-2206

Acknowledgements

On behalf of the study team, the authors would like to thank the patients and their families for participation in this study