1912

Inhibitory activity of TMC435350 on HCV NS3/4A proteases from genotypes 1 to 6

Corresponding author:
Tse-I Lin
Tibotec BVBA
Gen De Wittelaan L11 B3
2800 Mechelen, Belgium
+32/15.46.1436
tilin@its.jnj.com

Tse-I Lin[1], Benoit Devogelaere[1], Oliver Lenz[1], Origène Nyanguile[1], Elisabeth van der Helm[1], Katrien Vermeiren[1], Geneviève Vandercruyssen[1], Erna Cleiren[1], Jimmy Lindberg[2], Michael Edlund[2], Pierre Raboisson[1], Herman de Kock[1], Maxwell Cummings[1], Gregory Fanning[1], Kenneth Simmen[1]

[1]Tibotec BVBA, Mechelen, Belgium; [2]Medivir AB, Huddinge, Sweden

Introduction

Hepatitis C Virus (HCV; Figure 1):

- 120-170 million people infected;
- displays high degree of genetic variability;
- six genotypes;
- more than 50 subtypes;
- cure rates with current standard of care (pegylated interferon + ribavirin): ~40% for genotypes 1 and 4, ~80% for genotype 2, 3 and 6, and ~60% for genotype 5.[1,2]

Figure 1: Worldwide distribution of HCV genotypes 1 to 6.

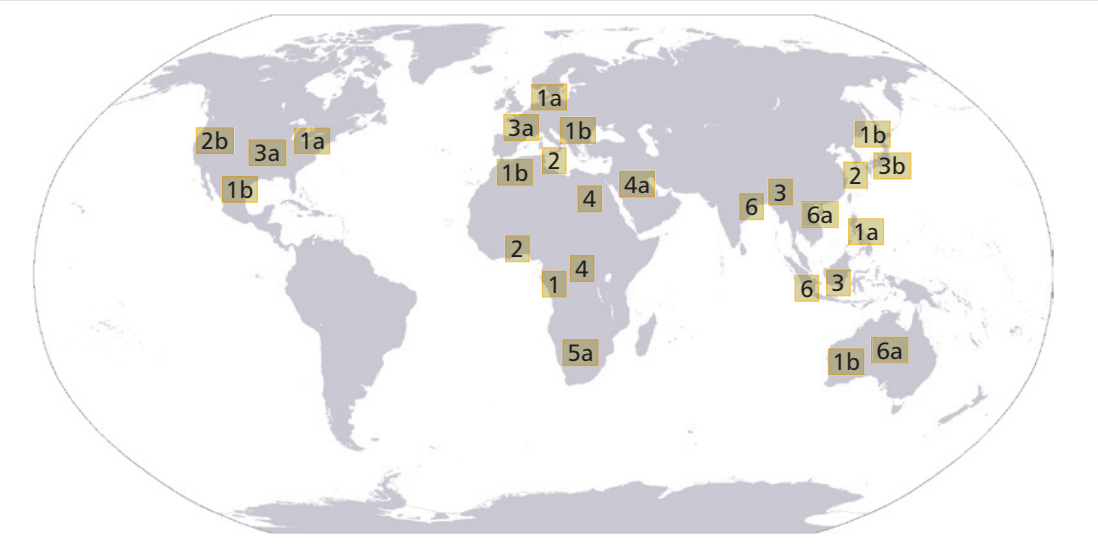
2b
3a
1a
1b
1a
3a
1b
1b
2
4
4a
6
3
6a
1b
2
3b
1a
2
1
4
6
3
5a
1b
6a

Based on references 2-4.

TMC435 (formerly known as TMC435350; Figure 2):

- Macrocyclic HCV NS3/4A protease inhibitor.
- EC_{50} = 8 nM on genotype 1b replicon.
- Limited effect of functional plasma protein binding (~2-fold shift of EC_{50} value).
- Currently in Phase 2a of clinical development.

Figure 2: Crystal structure of TMC435 bound to HCV NS3 protease. Surface representation of the enzyme, with part of the NS4A co-factor also visible (dark blue), and the inhibitor in color-by-atom stick format.

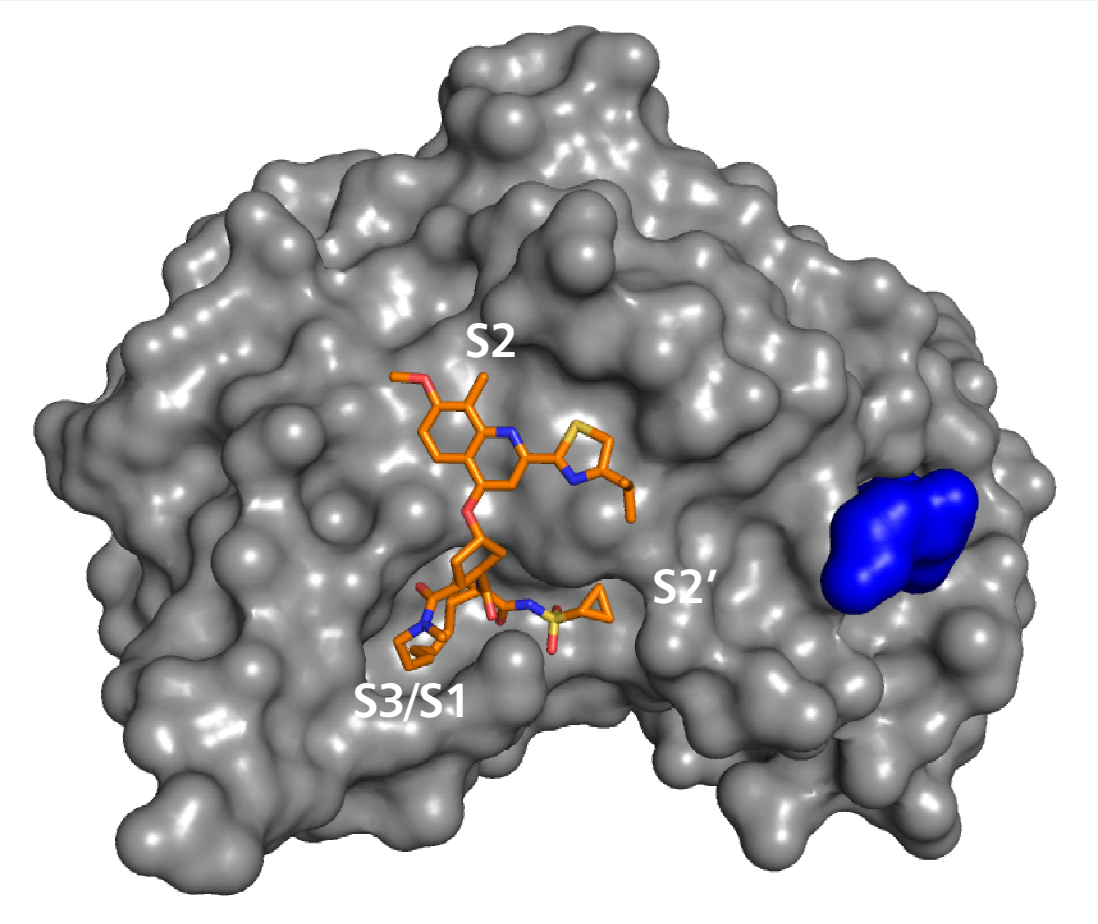
S2
S2′
S3/S1

The co-crystal structure of TMC435 bound to the protease domain of genotype 1b BK strain was determined at 2.4 Å resolution. The bound inhibitor spans S1′ (TMC435 cyclopropyl group) to S3 (TMC435 macrocycle). The methoxyquinoline group of TMC435 occupies the S2 subsite, which is relatively flat and (solvent) exposed.

Here we present the activity of TMC435 on a panel of genotype 1 to 6 NS3 enzymes obtained from patient isolates using biochemical protease assays and the kinetic binding analysis of genotype 1 isolates with surface plasmon resonance (SPR) based technology.

Results

Inhibitory activity of TMC435 against NS3 proteases of genotype 1 to 6

TMC435 activity on NS3 proteases derived from clinical isolates of genotypes 1a, 1b, 2b, 3a, 4a, 5a and 6a (Figure 3, Table 1):

- IC_{50} values for genotype 1a, 1b, 2b, 4a, 5a, and 6a proteases were less than 5-fold higher than the genotype 1b (con1) values.
- For genotype 3a protease, a ~10-fold reduction in inhibitory activity, compared to genotype 1, was observed.

Table 1: Inhibition of NS3 protease by TMC435 determined in a biochemical protease assay. Genotypes indicated in sample name as G1-G6.

genotype	1b				1a	
sample	G1b (con1)	G1b_03	G1b_05	G1b_08	G1a_07	G1a_08
Median IC_{50}(nM)	3.6	8.7	4.0	7.0	3.6	12.0
IQR [nM]	2.1-8.8	7.3-8.9	3.6-4.8	6.8-7.8	3.5-6.7	11.5-13.0

genotype	2b	3a	4a		5a	6a
sample	G2b	G3a	G4a	G4a_54	G5a_01	G6a_02
Median IC_{50}(nM)	1.9	36.5	8.9	4.6	6.2	2.3
IQR [nM]	0.6-3.1	32.8-51.5	0.8-9.9	4.3-5.0	6.1-7.2	2.2-2.4

IQR, interquartile range

NS3 proteases were cloned from clinical isolates of HCV genotype 1 to 6, expressed and purified from *E. coli*. The IC_{50} value for protease inhibition by TMC435 was determined by measuring the fluorescence resonance energy transfer (FRET) induced by the cleavage of the RetS1 peptide substrate when incubated with NS3 protease domain (supplemented with synthetic truncated NS4A peptide).

Figure 3: Inhibition of NS3 protease by TMC435 determined in a biochemical protease assay.

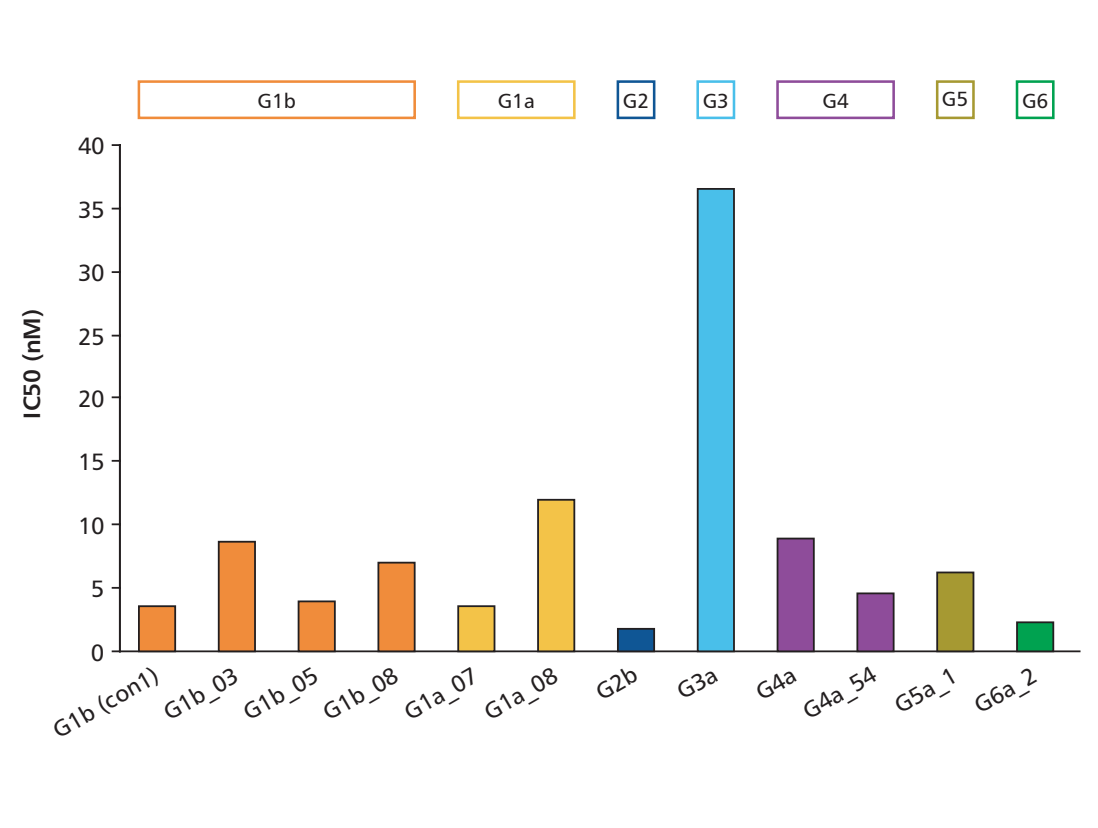
G1b
G1a
G2
G3
G4
G5
G6
IC50 (nM)
0
5
10
15
20
25
30
35
40
G1b (con1)
G1b_03
G1b_05
G1b_08
G1a_07
G1a_08
G2b
G3a
G4a
G4a_54
G5a_1
G6a_2

Genotype dependant difference of TMC435 binding pocket residues

Several residue positions in the region of the TMC435 binding site of NS3 show variation between different genotypes (Table 2, Figure 4A).

The reduced activity against genotype 3 enzymes can be explained by the predominant presence of D168Q variation in this genotype. Variations at position 168 can have a significant impact on the affinity of NS3 protease for TMC435 (Figure 4B).

- D168 plays a central role in a network of interactions that stabilizes the shape of the S2 pocket that is induced by TMC435 binding.
- Mutations of D168 will disturb this interaction network, through loss of stabilizing hydrogen bonds and/or by introduction of steric clashes.

Depending on the residue at position 168, significant alteration of the overall shape and chemical nature of the S2 region that binds the methoxyquinoline-group of TMC435 can be expected.
The D168E variation present in genotype 5a is conservative and hence allows preservation of intramolecular contacts.

Analysis of a single D168E or D168Q mutation in a genotype 1b background using a transient replicon assay confirmed the greater impact of D168Q on susceptibility to TMC435 compared to D168E, suggesting that the presence of D168Q in the genotype 3 protease might be pivotal for the reduced activity of TMC435 observed against the genotype 3 enzyme.

Table 2: Residue variation at the TMC435 binding site. Overview of the most predominant variations at positions in the TMC435 binding pocket with natural variation across genotypes 1-6. Analysis based on sequences deposited in Los Alamos and EU-HCV databases.

	Position								
genotype	78	79	80	122	123	132	155	156	168
1b	V	D	Q	S	R	V	R	A	D
1a	-	-	-	-	-	I	-	-	-
2a	A	E	G	K	-	L	-	-	-
2b	A	E	G	R	-	L	-	-	-
3a	-	-	-	-	T	L	-	-	Q
4a	-	-	-	T	-	I	-	-	-
5a	-	-	K	A	-	I	-	-	E
6a	-	-	K	N	-	I	-	-	-

REFERENCES
1. Strader DB et al. 2004. Hepatology 39: 1147.
2. Nguyen MH and Keeffe EB. 2005. Clin Gastroenterol Hepatol 3: S97.
3. Ramia S and Eid-Fares J. 2006. Int J Infect Dis 10: 272.
4. Simmonds P. 2004. J Gen Virol 85 (Pt 11): 3173-3188.

This poster will be available on-line at www.tibotec.com

Figure 4: Residue variation at the TMC435 binding site. (A) Crystal structure of TMC435 bound to HCV NS3 protease, showing residue positions in TMC435 binding pocket with natural variation between genotypes 1-6. The observed variable sites are in the vicinity of the S2 or S3 regions of the binding site. (B) Detailed view of the interactions between Q80, R155, D168, R123 and the methoxyquinoline group of TMC435.

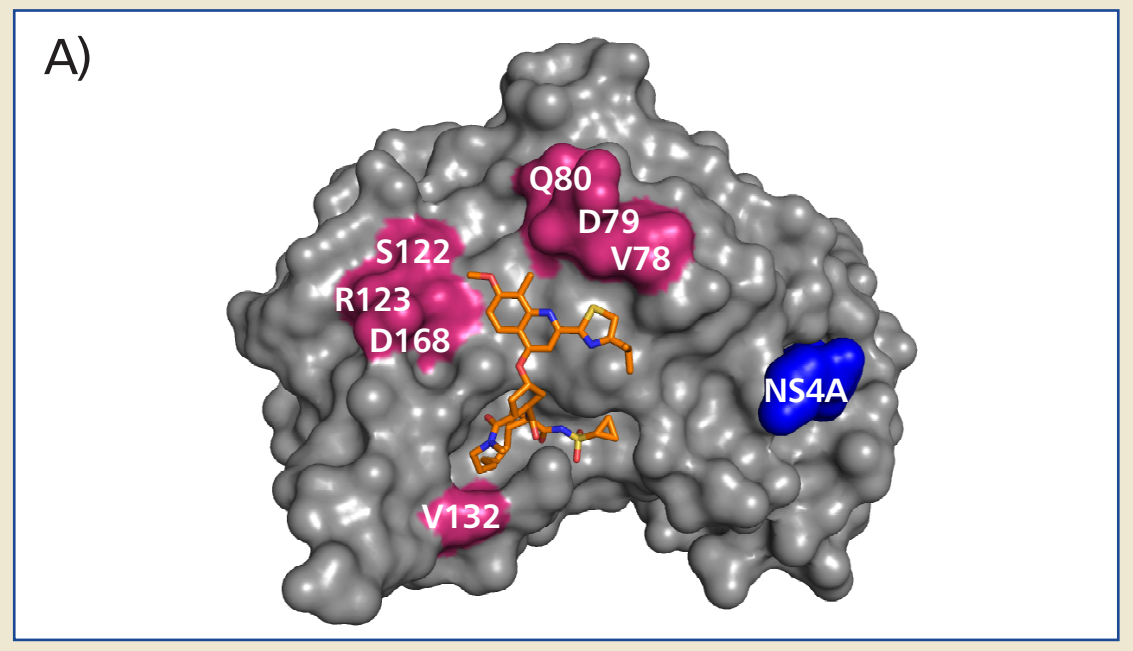
A)
Q80
D79
V78
S122
R123
D168
NS4A
V132

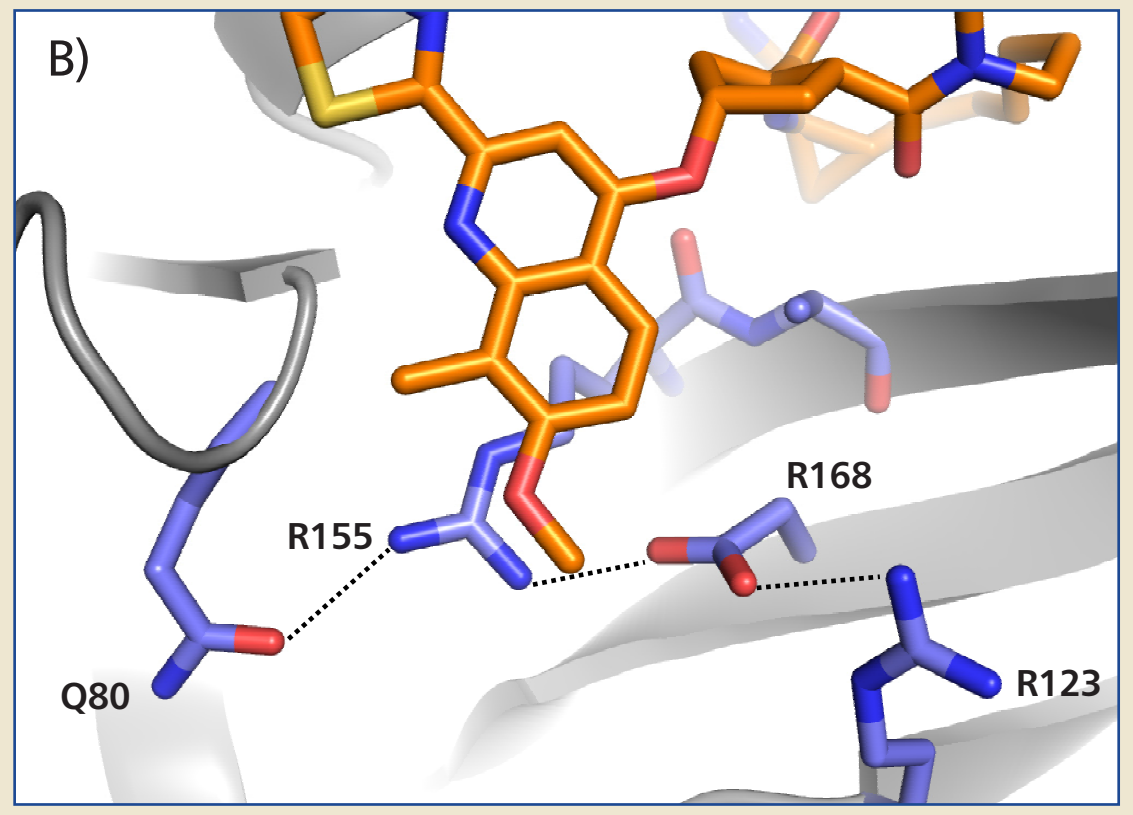
B)
R155
R168
Q80
R123

Binding of TMC435 to NS3 proteases of genotype 1

TMC435 binding to NS3 (Figure 5, Table 3) was consistent with:

- The similar IC_{50} values for genotype 1a and 1b samples (Figure 3).
- Non-covalent binding to the protease, with a fast association rate (k_{on} ~10^5 $M^{-1}s^{-1}$) and a slow dissociation rate (k_{off} ~10^{-3} s^{-1}). The dissociative half-life ($t_{1/2}$) of TMC435 is > 2 min.
- One-step binding kinetics (data not shown).

Figure 5: Binding of TMC435 to genotypes 1 NS3 protease determined by SPR. (A) Experimental sensorgram showing the interaction between TMC435 (concentration of TMC435 used: 15 to 792 nM) and genotype 1b (con1) NS3. (B) Interaction kinetic map of genotypes 1 NS3 proteases with TMC435, shown as association versus dissociation rate constants (k_{on} and k_{off}), and the combinations of k_{on} and k_{off} that result in the same K_D values (diagonal lines).

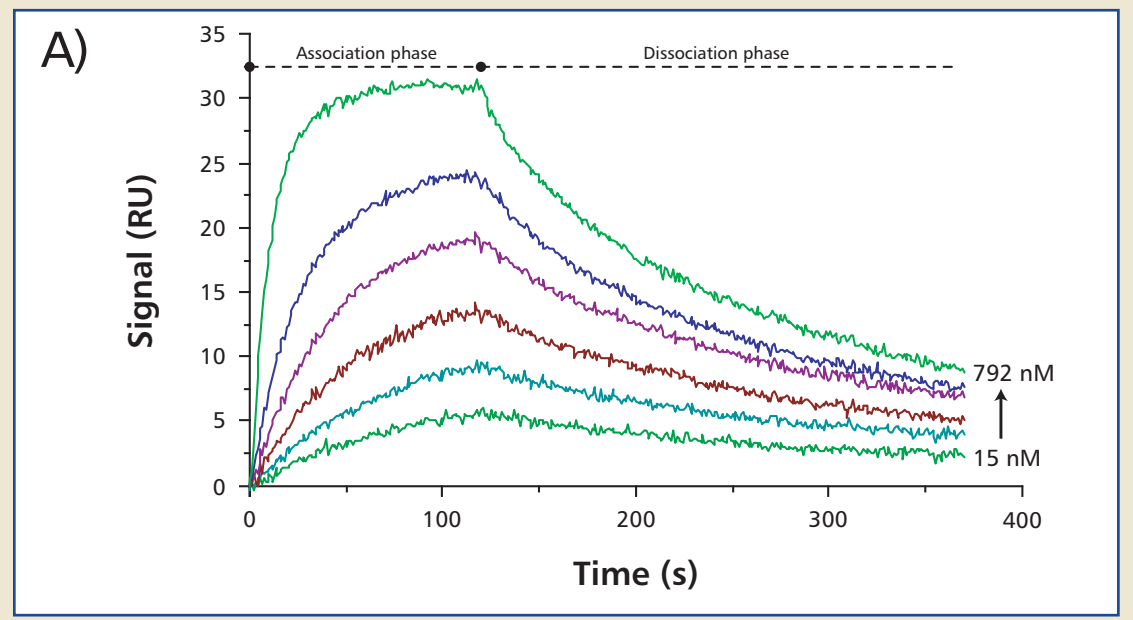
A)
Association phase
Dissociation phase
Signal (RU)
0
5
10
15
20
25
30
35
792 nM
15 nM
0
100
200
300
400
Time (s)

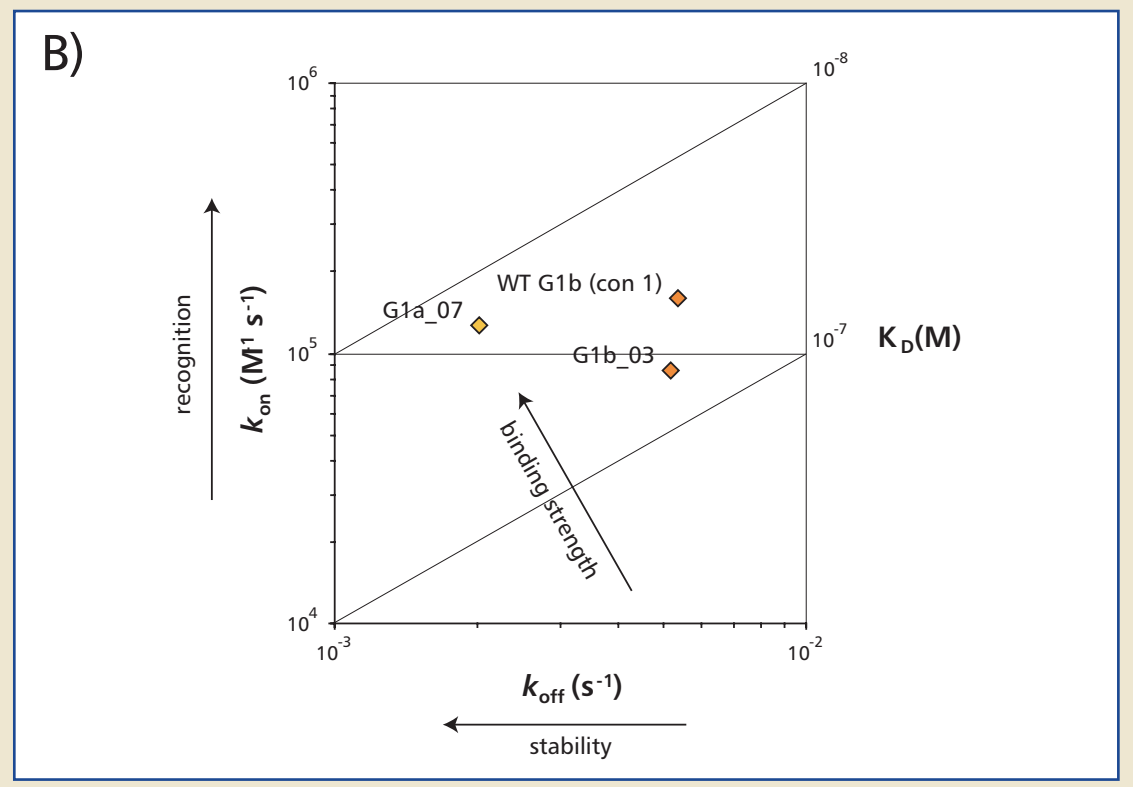
B)
recognition
k_{on} (M^{-1} s^{-1})
10^6
10^5
10^4
10^{-8}
10^{-7}
K_D(M)
G1a_07
WT G1b (con 1)
G1b_03
binding strength
10^{-3}
10^{-2}
k_{off} (s^{-1})
stability

NS3 proteases were cloned from genotype 1 clinical isolates or from strain con1b, expressed and purified from *E. coli*. To characterize the binding of TMC435 to HCV protease, binding kinetics to immobilized recombinant genotype 1 NS3 protease domain was determined using SPR on a Biacore S51 (GE Healthcare).

Table 3: Parameters of binding of TMC435 to genotypes 1 NS3 protease determined by SPR.

genotype	1a	1b	
sample	G1a_07	G1b_03	WT G1b
K_D (M)	1.61×10^{-8}	5.97×10^{-8}	3.35×10^{-8}
k_{off} (s^{-1})	2.03×10^{-3}	5.17×10^{-3}	5.36×10^{-3}
k_{on} ($M^{-1}s^{-1}$)	1.26×10^{5}	8.66×10^{4}	1.60×10^{5}
$t_{1/2}$ (s)	341	134	129

Conclusion

- TMC435 is a potent inhibitor of NS3/4A protease from genotype 1 to 6, with IC_{50} values below 13 nM for all HCV NS3/4A enzymes tested with the exception of genotype 3a protease (37 nM).
- Analysis suggests that residue diversity at D168 is pivotal for reduced activity of TMC435 observed against genotype 3a protease.
- TMC435 binds to the protease non-covalently, with a fast association rate and a slow dissociation rate. The dissociative half-life ($t_{1/2}$) of TMC435 is > 2 min.
- *In vitro* potency, in conjunction with its pharmacokinetic properties in humans, suggests that TMC435 may provide benefits as an antiviral agent for individuals infected with non-genotype-1 HCV.

59th Annual Meeting of the American Association for the Study of Liver Diseases, October 31-November 4, 2008, San Francisco, CA, United States