The Topical Application of Acyclovir Five Percent and Hydrocortisone One Percent Cream in Subjects with Recurrent Herpes Labialis is Not Associated with Acyclovir Resistance

Mia Brytting, PhD[1], Eva Arlander, PhD[2], Larry N. Gever, PharmD[3] and William Wheeler, PhD[3]

[1]*Swedish Institute of Infectious Disease Control, Stockholm, Sweden;* [2]*Medivir AB, Huddinge, Sweden;* [3]*Meda Pharmaceuticals, Inc., Somerset, NJ*

ABSTRACT

Purpose: The combination of acyclovir five percent and hydrocortisone one percent cream (Xerese, ACH) is approved for the topical treatment of recurrent herpes labialis. A summary of virology assessments performed in the ACH phase three clinical trial program are reported here.

Methods: Informed consent was obtained for all subjects and the ethics committees/institutional review boards approved the studies. The Phase three program comprised of two trials (609-04 immunocompetent subjects; 609-06 immunocompromised subjects). Virology assessments performed in these trials included viral sampling, viral isolation, acyclovir (AC) susceptibility studied by plaque reduction assay (PRA), and thymidine kinase (TK) and DNA polymerase genotyping.

Results: A total of 1550 subjects (609-04, n equals 1443; 609-06, n equals 107) received study medication in the phase three program and were included in the intention-to-treat population. For study 609-04, 32 (22 percent) of subjects in the ACH group, 44 (24 percent) of subjects in the acyclovir (AC) group, and 31 (40 percent) of subjects in the vehicle group had ulcerative recurrences with positive virus samples collected. In the ACH, AC, and vehicle groups, 18, 29, and 24 subjects had a positive viral culture and a healing time longer than 5.5 days, respectively. All samples were ACV sensitive and no mutations inducing ACV resistance were determined, including samples from subjects with a prolonged healing time. For study 609-06, positive quantitative PCR samples were obtained from 22 (76 percent) subjects receiving ACH and 8 (67 percent) subjects receiving AC. In the ACH and AC groups, 13 (45 percent) and 7 (58 percent) subjects had ulcerative recurrences with positive virus samples collected, respectively. None of the subjects had reduced AC sensitivity due to a mutation in the TK or DNA-polymerase genes. No AC resistance was identified in any subject.

Conclusion: Virology assessments from the ACH phase three program found no AC resistance in any subject. Topical treatment with hydrocortisone in addition to AC did not promote the emergence of resistant virus strains.

INTRODUCTION

- Herpes simplex virus (HSV) infections are treated with antiviral drugs such as acyclovir (ACV).[1] Herpes labialis (cold sore, fever blister) is the most common clinical presentation of recurrent HSV-1 infection.[2]
- AC resistance frequency is low (≤0.6%) among immunocompetent patients.[1] The majority of cases are found in viral isolates from immunocompromised subjects, ranging from 3–6% and up to 14% in bone marrow transplant patients.[1] AC resistance is associated with mutations in the TK and DNA-polymerase genes.[1]
- Acyclovir 5% and hydrocortisone 1% cream (AHC) is approved as Xerese™ for the treatment of recurrent herpes labialis (cold sores) in the United States and as Xerclear™ in the European Union.
- Topical application of AHC has been shown to be safe and effective in the treatment of recurrent herpes labialis.[3]

OBJECTIVES

The objectives of these studies were to evaluate the:

- Efficacy and tolerability of AHC in immunocompetent and immunocompromised adults aged ≥18 years with recurrent herpes labialis;
- This report represents a summary of all of the virology assessments performed in the Phase 3 program for AHC.

METHODS

Study Design

- Study 609-04: A Phase 3, multicenter, randomized, double-blind, parallel, vehicle-controlled study in immunocompetent adults with recurrent herpes simplex labialis. Subjects received topical AHC, AC (in AHC vehicle) or vehicle, 5 times a day for 5 days.
- Study 609-06: A Phase 3, multicenter, randomized, double-blind, parallel, active-controlled study in immunocompromised adults with recurrent herpes simplex labialis. Subjects received topical AHC or AC (in AHC vehicle), 5 times a day for 5 days.

Viral Sampling

- Obtained at study visits from subjects with ulcerative recurrences.
- Swabs were collected from lesions at the ulcer/soft crust stages to avoid interference with the healing process.

Virology Assessments

- *Study 609-04*
 - Viral isolation (culturing) was conducted for all collected samples.
 - Positive samples from subjects with a prolonged healing time were assessed for AC susceptibility according to the US antiviral susceptibility testing procedure for HSV, plaque reduction assay (PRA).
 - Genotyping of the TK gene in some subjects was also performed.
- *Study 609-06*
 - Quantitative polymerase chain reaction (PCR) and virus isolation were conducted for all collected samples and were analyzed for AC susceptibility.
 - AC resistant strains were further tested with regards to AC susceptibility in the presence of hydrocortisone.
 - Susceptibility testing also included genotyping, including PCR and subsequent sequencing of the genes coding for TK- and DNA polymerase.

RESULTS

Study Disposition and Demographics

- Study 609-04: 1443 immunocompetent subjects were enrolled in 51 study sites in the US and 4 in Canada (72% females; mean age 44 years).
- Study 609-06: 107 immunocompromised subjects were enrolled in 19 study sites in Russia and 6 in the Ukraine (46% females; mean age 32 years).

Virological Assessments

Study 609-04

- Of the 403 immunocompetent subjects with ulcerative recurrences out of 1443 [intention-to-treat (ITT) population] who were sampled for virus isolation, 107 subjects had HSV culture positive samples (22%, 24%, and 40% for the AHC, AC, and vehicle treated groups, respectively [**Figure 1**]).

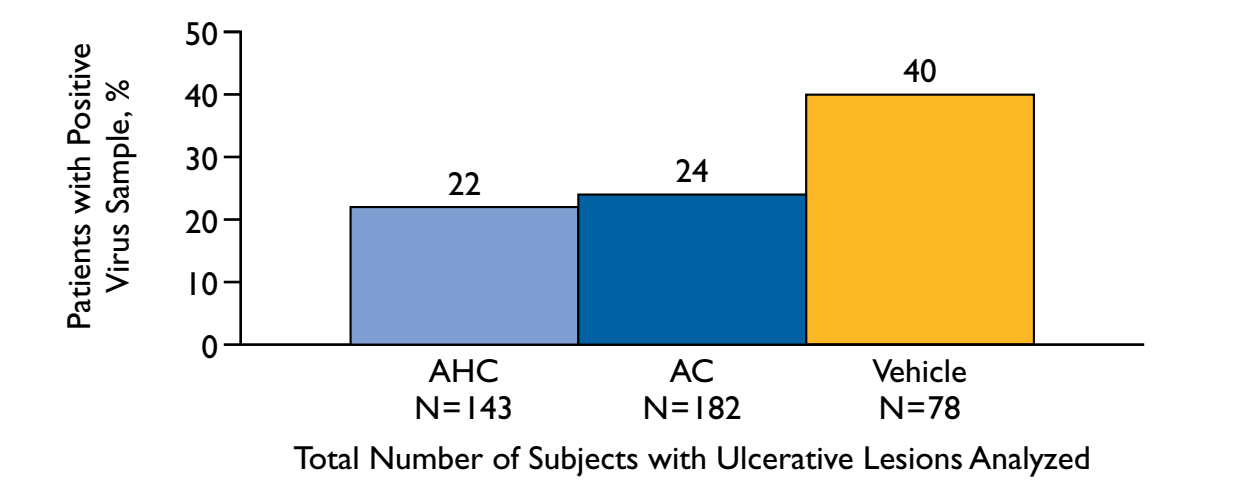

Patients with Positive Virus Sample, %
50
40
30
20
10
0
22
24
40
AHC N=143
AC N=182
Vehicle N=78
Total Number of Subjects with Ulcerative Lesions Analyzed

- In total, 71 out of 107 immunocompetent subjects with HSV culture positive sample had a healing time longer than 5.5 days. The percentages were 56% for AHC, 66% for AC, and 77% for the vehicle treated group (**Figure 2**).

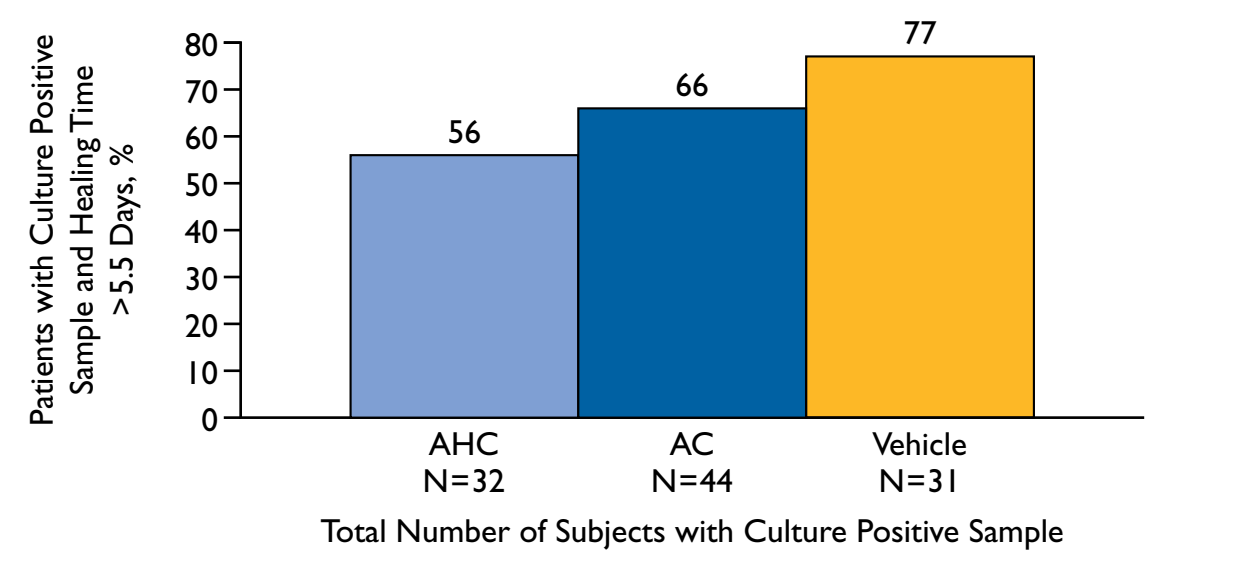

Patients with Culture Positive Sample and Healing Time >5.5 Days, %
80
70
60
50
40
30
20
10
0
56
66
77
AHC N=32
AC N=44
Vehicle N=31
Total Number of Subjects with Culture Positive Sample

Study 609-06

- In total, 41 immunocompromised subjects with ulcerative recurrences out of 107 subjects in the ITT population were sampled for virus isolation. Out of them, 20 subjects had HSV culture positive samples, 45% and 58% in the AHC and AC treated groups, respectively (**Figure 3**).

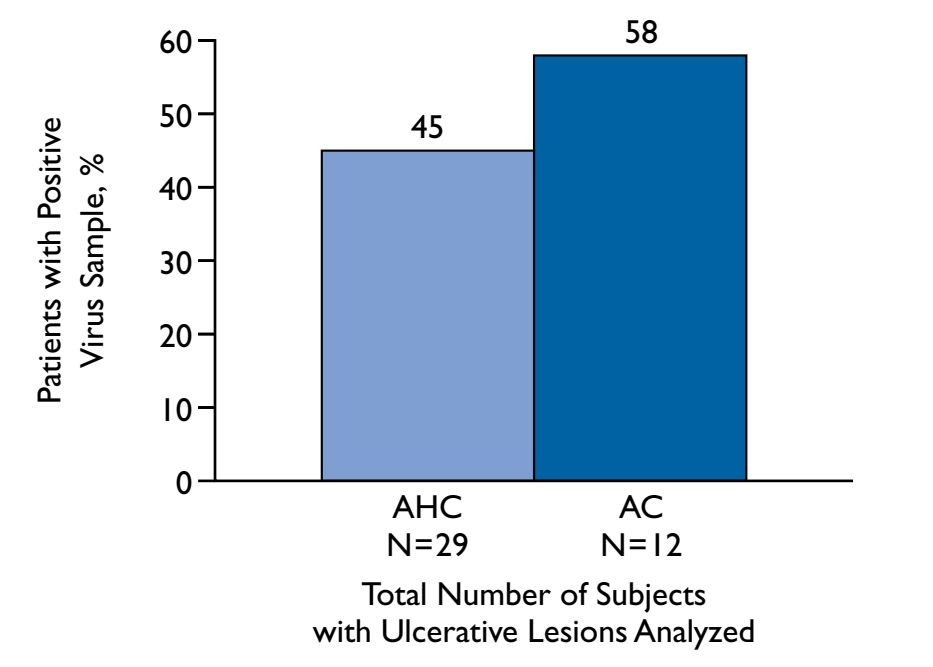

Patients with Positive Virus Sample, %
60
50
40
30
20
10
0
45
58
AHC N=29
AC N=12
Total Number of Subjects with Ulcerative Lesions Analyzed

- Of the 41/107 immunocompromised subjects with ulcerative recurrences in the ITT population who were sampled for virus isolation, 30 subjects had HSV DNA positive samples analyzed by quantitative PCR. The percentages were 76% and 67% for the AHC and AC treated groups, respectively (**Figure 4**).

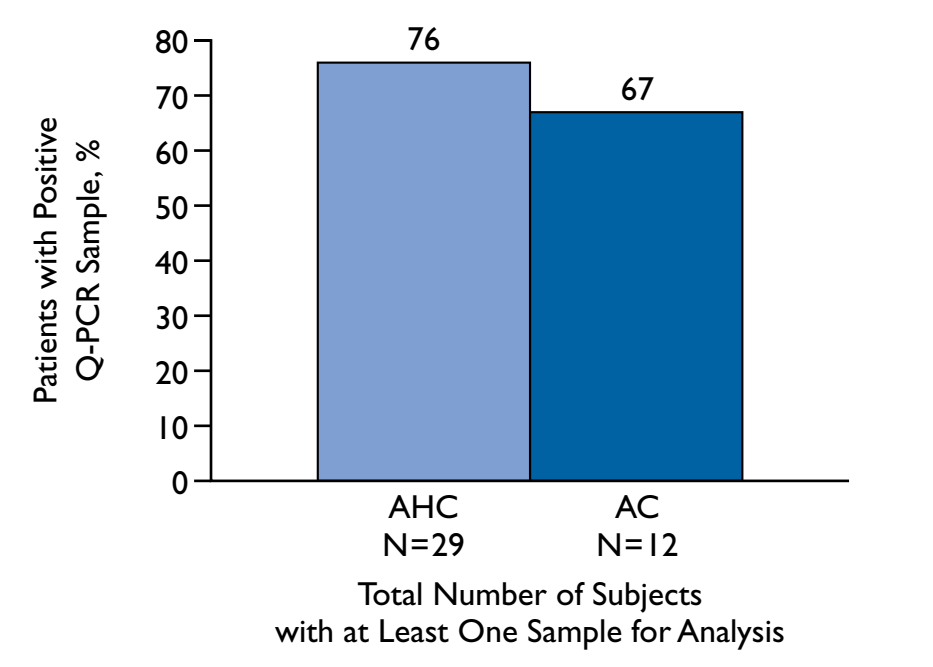

Patients with Positive Q-PCR Sample, %
80
70
60
50
40
30
20
10
0
76
67
AHC N=29
AC N=12
Total Number of Subjects with at Least One Sample for Analysis

Acyclovir Susceptibility

- In Study 609-04, all the 71 subjects with HSV positive isolates had acyclovir sensitive strains characterized by phenotypic (PRA) and/or genotypic (TK sequencing) assays.
- In Study 609-06, all the 20 subjects with HSV positive isolates and 6 of the subjects with only DNA positive samples had acyclovir sensitive strains characterized by phenotypic (PRA) and/or genotypic (TK and DNA-polymerase sequencing) assays. Additional samples from two subjects with only HSV DNA positive samples were characterized with TK sequencing and no mutation known to induce acyclovir resistance was determined. The last two subjects with only HSV DNA positive samples had too low viral load, so the samples could not be characterized.

REFERENCES

1. Morfin F, Thouvenot D. Herpes simplex virus resistance to antiviral drugs. *Journal of Clinical Virology.* 2003;26(1):29-37.
2. Stanberry LR, Oxman MN, Simmons A. Herpes Simplex Viruses In: Gorbach SL, Bartlett JG, Blacklow NR, eds. *Infectious Diseases*. Philadelphia: Lippincott Williams & Wilkins; 2004:1905-1917.
3. Hull C, Harmenberg J, Arlander E, et al. Early treatment of cold sores with topical ME-609 decreases the frequency of ulcerative lesions: A randomized, double-blind, placebo-controlled, patient-initiated clinical trial. *J Am Acad Dermatol*. 2010; Sep 17. [Epub ahead of print]

CONCLUSIONS

- **No acyclovir resistance was identified in HSV isolates from immunocompetent and immunocompromised subjects in the AHC Phase 3 program.**
- **Topical administration of hydrocortisone with AC did not promote the emergence of HSV strains resistant to AC in immunocompetent and immunocompromised subjects with recurrent herpes labialis.**

This research was funded by Medivir AB.