Poster 1075

Pharmacokinetic-pharmacodynamic (PK-PD) analyses of TMC435 in treatment-naive hepatitis C (HCV)-infected patients in the OPERA-1 study

V. Sekar[1], P. Vis[2], O. Lenz[2], P. Meyvisch[2], M. Peeters[2], G. De Smedt[2]

[1]Tibotec Inc, USA; [2]Tibotec BVBA, Belgium

Corresponding author:
Vanitha Sekar
Tibotec Inc,
1125 Trenton-Harborton Road,
Titusville, New Jersey 08560, USA
+1 609-730-7521
e-mail vsekar@its.jnj.com

Introduction

- **TMC435 is a macrocyclic NS3/4A protease inhibitor currently in Phase IIb clinical development for the treatment of hepatitis C virus (HCV) infection.**
- **It is a potent and selective inhibitor of NS3/4A *in vitro*, with a 50% effective concentration (EC_{50}) of 8 nM in a genotype-1b replicon cell line.[1]**
- **Findings from Phase I and IIa studies have shown that TMC435 is well tolerated and has a pharmacokinetic (PK) profile that supports a once-daily (QD) dosing regimen.[2-5]**
- **Optimal Protease inhibitor Enhancement of Response to TherApy (OPERA)-1 is an ongoing randomised, double-blind, placebo-controlled, Phase IIa, proof-of-concept study.**
- **A 4-week analysis has shown that TMC435 demonstrates potent antiviral activity in both treatment-naive and -experienced patients infected with HCV genotype-1.[3-5]**
- **Here we describe the pharmacokinetic-pharmacodynamic (PK-PD) relationships for efficacy and safety markers in treatment-naive HCV-infected patients who received TMC435 in the OPERA-1 study.**

Methods

Study design

- OPERA-1 (TMC435-C201; NCT00561353) is a randomised, double-blind, placebo-controlled, Phase IIa proof-of-concept study (Figure 1).

Figure 1. Study design of OPERA-1.

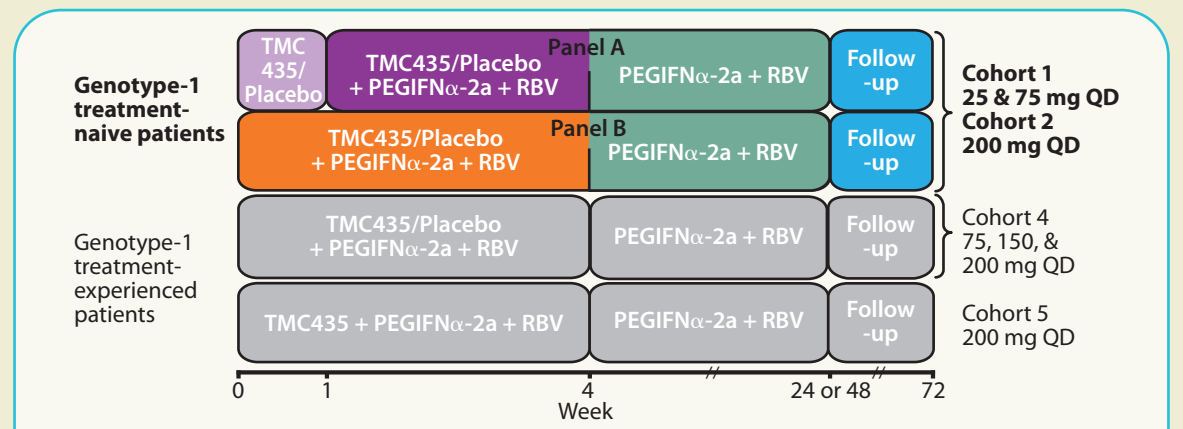

Genotype-1 treatment-naive patients
Genotype-1 treatment-experienced patients
TMC 435/ Placebo
Panel A
TMC435/Placebo + PEGIFNα-2a + RBV
PEGIFNα-2a + RBV
Follow -up
Panel B
TMC435/Placebo + PEGIFNα-2a + RBV
PEGIFNα-2a + RBV
Follow -up
TMC435/Placebo + PEGIFNα-2a + RBV
PEGIFNα-2a + RBV
Follow -up
TMC435 + PEGIFNα-2a + RBV
PEGIFNα-2a + RBV
Follow -up
Cohort 1 25 & 75 mg QD
Cohort 2 200 mg QD
Cohort 4 75, 150, & 200 mg QD
Cohort 5 200 mg QD
0
1
4
24 or 48
72
Week

PEGIFNα-2a, pegylated interferon alfa-2a; RBV, ribavirin; QD, once daily

- Eligible patients were aged 18–70 years with documented chronic genotype-1 HCV infection, plasma HCV ribonucleic acid (RNA) levels of ⩾10,000 IU/mL at screening and no symptoms of decompensated liver disease.
- Treatment-naive patients were enrolled into Cohorts 1 and 2.
- In Cohort 1, patients received TMC435 at 25 mg or 75 mg QD, or placebo; in Cohort 2, patients received TMC435 at 200 mg QD or placebo.
- In Cohorts 1 and 2, patients received TMC435 either as 7 days of monotherapy followed by 21 days of triple therapy (i.e. TMC435 or placebo combined with pegylated interferon-alfa-2a [P] 180 µg once weekly [QW] plus ribavirin [R] 1,000–1,200 mg twice daily [BID] dependent on body weight) (Panel A), or as 28 days of triple therapy (Panel B).
- Following the 28-day TMC435/placebo treatment period, patients continued to receive P/R up to Week 48.

Pharmacokinetic assessments and analysis methods

- PK profiles of TMC435 were determined on Days 7 and 28 of treatment, i.e. following monotherapy and triple therapy, respectively.
 - PK analysis was performed using non-compartmental methods using the WinNonlin Professional™ (Version 4.1; Pharsight Corporation, Mountain View, California, USA).
- Calculated PK parameters included:
 - time to reach the maximum plasma concentration (t_{max})
 - maximum plasma concentration (C_{max})
 - minimum plasma concentration (C_{min})
 - pre-dose plasma concentration (C_{0h})
 - area under the concentration-time curve (AUC) from time of administration up to 24 hours post-dosing (AUC_{24h}).
- The relationship between TMC435 PK and antiviral activity at Day 28 was explored by evaluating change from baseline to Day 28 in HCV RNA levels.
 - HCV RNA levels were determined using the TaqMan HCV/HPS assay v2.0, which has a dynamic range of 25–391,000,000 IU/mL with a lower limit of quantification of 25 IU/mL.
- PK-PD assessments for safety at Day 28 included change from baseline in alkaline phosphatase (ALP), aspartate aminotransferase (AST), alanine aminotransferase (ALT) and serum bilirubin.

- PK simulations were performed to predict TMC435 exposure at different doses based on a population PK model developed using data from the first in-human study (TMC435-C101)[2] and OPERA-1 in treatment-naive genotype-1 HCV-infected patients who received TMC435 at 25, 75 or 200 mg QD.
 - The population PK model was developed using PK data after repeated administration.
 - Clearance was modelled to decrease in a non-linear fashion with increasing dose, and the absorption rate constant was modelled to increase linearly with an increasing dose.
 - The model performed well based on goodness-of-fit and was considered adequate to perform simulations to predict TMC435 exposure.
- PK simulations are presented here to describe the rationale for the doses selected for further evaluation in the ongoing Phase IIb **P**rotease **I**nhibitor TMC435 trial assessing the optima**L** dose and duration as once dai**L**y **A**nti-viral **R**egimen (PILLAR; NCT00882908).

Results

Subject disposition

- In Cohorts 1 and 2, 74 patients were included in the intent-to-treat (ITT) population.
 - Fifty-five patients received TMC435 and 19 received placebo.
- Patient demographics and baseline characteristics were well balanced between treatment groups in both cohorts (Table 1).

Table 1. Patient demographics and baseline characteristics of patients included in Cohorts 1 and 2 of OPERA-1.

	Cohort 1 TMC435 25 mg N=18	Cohort 1 TMC435 75 mg N=19	Cohort 1 placebo N=13	Cohort 2 TMC435 200 mg N=18	Cohort 2 placebo N=6	Total N=74
Gender						
Female	5 (27.8)	8 (42.1)	3 (23.1)	8 (44.4)	1 (16.7)	25 (33.8)
Male	13 (72.2)	11 (57.9)	10 (76.9)	10 (55.6)	5 (83.3)	49 (66.2)
Race						
Black	1 (5.6)	0	0	2 (11.1)	0	3 (4.1)
Caucasian/white	17 (94.4)	19 (100)	12 (92.3)	16 (88.9)	6 (100)	70 (94.6)
Other	0	0	1 (7.7)	0	0	1 (1.4)
Age (years)						
Median (range)	52.0 (22-64)	47.0 (22-70)	45.0 (19-60)	46.5 (19-68)	44.5 (19-50)	47.0 (19-70)
Category						
⩽40	3 (16.7)	7 (36.8)	5 (38.5)	6 (33.3)	2 (33.3)	23 (31.1)
41-54	9 (50.0)	7 (36.8)	5 (38.5)	10 (55.6)	4 (66.7)	35 (47.3)
>54	6 (33.3)	5 (26.3)	3 (23.1)	2 (11.1)	0	16 (21.6)
Body mass index						
Median (range)	27.4 (23-32)	25.0 (18-32)	25.7 (19-32)	23.7 (19-31)	25.9 (20-32)	25.0 (18-32)
Cirrhosis						
With cirrhosis	11 (61.1)	9 (47.4)	7 (53.8)	3 (16.7)	2 (33.3)	32 (43.2)
HCV RNA						
Median log value (range)	6.7 (5-7)	6.4 (5-8)	6.6 (6-7)	6.6 (5-7)	6.4 (5-7)	6.6 (5-8)
Category (IU/mL)						
<800,000	1 (5.6)	5 (26.3)	2 (15.4)	2 (11.1)	2 (33.3)	12 (16.2)
⩾800,000	17 (94.4)	14 (73.7)	11 (84.6)	16 (88.9)	4 (66.7)	62 (83.8)
HCV subtype (NS5B)[a]						
1	0	0	0	1 (5.6)	0	1 (1.4)
1a	6 (33.3)	7 (36.8)	7 (58.3)	5 (27.8)	5 (83.3)	30 (41.1)
1b	11 (61.1)	12 (63.2)	5 (41.7)	12 (66.7)	1 (16.7)	41 (56.2)
1e	1 (5.6)	0	0	0	0	1 (1.4)
ALT toxicity grade						
Normal range	8 (44.4)	5 (26.3)	5 (38.5)	8 (44.4)	2 (33.3)	28 (37.8)
Grade 1	7 (38.9)	9 (47.4)	3 (30.8)	7 (38.9)	3 (50.0)	30 (40.5)
Grade 2	1 (5.6)	2 (10.5)	2 (15.4)	3 (16.7)	1 (16.7)	9 (12.2)
Grade 3	1 (5.6)	2 (10.5)	2 (15.4)	0	0	5 (6.8)
Grade 4	1 (5.6)	1 (5.3)	0	0	0	2 (2.7)

[a]For 1 subject in the placebo group of Cohort 1 the HCV subtype (NS5B) was unknown
ALT, alanine aminotransferase; HCV, hepatitis C virus; RNA, ribonucleic acid

Pharmacokinetic profile of TMC435

- The PK profile of TMC435 on Days 7 (monotherapy group [Panel A]) and 28 (monotherapy and triple therapy groups [Panels A and B]) are shown in Table 2.
- Steady-state conditions were reached by Day 7 for TMC435.
- Steady-state PK parameters for TMC435 were comparable following TMC435 monotherapy (Day 7) and triple therapy (Day 28), indicating a lack of effect of P/R co-administration on TMC435 exposure.

Table 2. Pharmacokinetic profile of TMC435 in treatment-naive patients in Cohorts 1 and 2 of OPERA-1 who received 7 days of TMC435 monotherapy followed by 21 days of triple therapy (TMC435+P/R) (A) or 28 days of triple therapy (TCM435+P/R) (B).

(A)

Pharmacokinetics of TMC435 (median [range])	Panel A: TMC435 25 mg QD	Panel A: TMC435 75 mg QD	Panel A: TMC435 200 mg QD
Day 7			
n	9	9	7[a]
C_{0h}, ng/mL	57.8 (13.6-148)	420 (89.7-860)	2,770 (929-11,000)
C_{min}, ng/mL	48.5 (13.6-145)	392 (86.7-786)	2,240 (850-9,730)
C_{max}, ng/mL	262 (162-542)	1,410 (597-2,450)	6,770 (4,140-18,600)
t_{max}, h	6.02 (2.08-8.00)	6.00 (4.00-6.03)	6.00 (4.00-6.00)
AUC_{24h}, ng.h/mL	3,326 (1,989-8,518)	20,080 (6,314-33,650)	119,700 (48,560-332,900)
	Panel A: TMC435 25 mg QD + P/R	**Panel A: TMC435 75 mg QD + P/R**	**Panel A: TMC435 200 mg QD + P/R**
Day 28			
n	9	8[b]	7[c]
C_{0h}, ng/mL	62.6 (17.1-107)	163 (101-991)	3,560 (512-19,400)
C_{min}, ng/mL	60.8 (17.1-107)	163 (95.3-794)	3,150 (505-17,500)
C_{max}, ng/mL	267 (201-432)	897 (508-1,930)	7,410 (3,490-24,800)
t_{max}, h	4.07 (3.98-10.00)	6.00 (4.00-6.03)	6.04 (4.00-10.08)
AUC_{24h}, ng.h/mL	3,381 (2,350-6,852)	11,620 (7,479-33,490)	112,500 (43,850-504,300)

[a]n=8 for C_{0h}, C_{min}
[b]n=9 for C_{0h}, C_{min}
[c]n=8 for C_{0h}, C_{min}, C_{max} and t_{max}

(B)

Pharmacokinetics of TMC435 (median [range])	Panel B: TMC435 25 mg QD + P/R	Panel B: TMC435 75 mg QD + P/R	Panel B: TMC435 200 mg QD + P/R
Day 28			
n	9	9	10
C_{0h}, ng/mL	75.8 (21.2-221)	297 (77.2-3,610)	3,550 (113-14,200)
C_{min}, ng/mL	65.4 (21.2-221)	294 (62.2-3,090)	3,260 (111-12,600)
C_{max}, ng/mL	226 (144-677)	1,160 (395-4,790)	9,090 (3,740-23,600)
t_{max}, h	5.92 (4.00-6.05)	6.00 (3.87-8.00)	6.00 (4.00-8.00)
AUC_{24h}, ng.h/mL	3,080 (2,170-10,470)	16,160 (4,926-92,720)	150,500 (34,120-395,800)

AUC_{24h}, area under the concentration-time curve from time of administration up to 24 hours; C_{0h}, pre-dose plasma concentration; C_{max}, maximum plasma concentration; C_{min}, minimum plasma concentration; P, pegylated interferon alfa-2a; QD, once daily; R, ribavirin; t_{max}, time to reach the maximum plasma concentration

- A dose-proportional increase in TMC435 plasma concentrations was observed across the 25–75 mg dose range. However, the increase in TMC435 plasma concentrations observed across the 75–200 mg dose range was more than dose-proportional.
 - The difference in AUC_{24h} between TMC435 at 75 and 200 mg (a 2.7-fold increase in dose) was 8-fold.
- Following treatment with TMC435 doses of 25, 75 and 200 mg QD, mean trough TMC435 concentrations at Day 28 were approximately 10-fold, 50-fold and 800-fold higher than the EC_{50} value (6 ng/mL), respectively.

PK-PD and antiviral activity

- Mean change from baseline to Day 28 in plasma HCV RNA levels were -4.26, -4.48 and -4.61 $\log_{10}$ IU/mL in patients who received initial monotherapy with TMC435 at 25, 75 and 200 mg QD, respectively, followed by 21 days of triple therapy; and -4.74, -5.52 and -5.44 $\log_{10}$ IU/mL in patients who received 28 days of triple therapy with TMC435 at 25, 75 and 200 mg QD, respectively.[5]
- Change from baseline to Day 28 in plasma HCV RNA ($\log_{10}$ IU/mL) versus TMC435 exposure (defined as AUC_{24h}) is shown in Figure 2.

Figure 2. Change in $\log_{10}$ plasma HCV RNA from baseline to Day 28 as a function of TMC435 exposure (defined as AUC_{24h}).

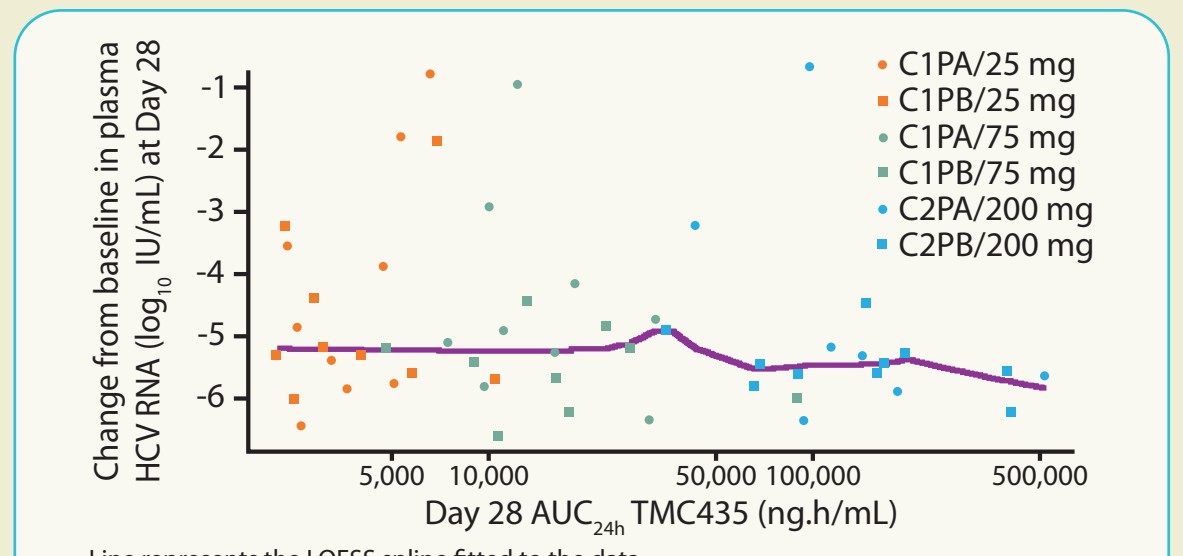

Change from baseline in plasma HCV RNA (log10 IU/mL) at Day 28
-1
-2
-3
-4
-5
-6
C1PA/25 mg
C1PB/25 mg
C1PA/75 mg
C1PB/75 mg
C2PA/200 mg
C2PB/200 mg
5,000 10,000
50,000 100,000
500,000
Day 28 AUC24h TMC435 (ng.h/mL)

Line represents the LOESS spline fitted to the data
AUC, area under the concentration-time curve; C, cohort; HCV, hepatitis C virus; PA, panel A; PB, panel B; RNA, ribonucleic acid

- No clear PK-PD relationship was observed with TMC435 at 75 mg QD doses or above.

PK-PD and safety

- Change from baseline to Day 28 in total, direct and indirect bilirubin; ALP; AST and ALT levels as a function of TMC435 exposure (defined as AUC_{24h}) are shown in Figure 3.
- A trend towards mild increases in serum bilirubin and ALP was observed with increasing TMC435 exposure, with increases mainly observed at the 200 mg dose.
- No consistent PK-PD relationship was observed for other liver parameters.

Population PK modelling

- The simulated ratios of the medians for TMC435 PK parameters are shown in Table 3.
- Using TMC435 at 25 mg QD as a reference, TMC435 at 75, 150 and 200 mg QD demonstrated non-linear PK after multiple dosing.

Table 3. Simulated ratios of the medians of TMC435 pharmacokinetic parameters.

Ratio of medians (TMC435 25 mg QD as reference dose)	TMC435 dose (mg, given QD) 25	75	100	150	200
C_{max}	1	4.1	6.9	16	32
AUC	1	4.1	7	18	40
C_{0h}	1	4	7.5	22	62
C_{min}	1	3.9	7.7	23	63

AUC, area under the concentration-time curve; C_{0h}, pre-dose plasma concentration; C_{max}, maximum plasma concentration; C_{min}, minimum plasma concentration; QD, once daily

- When comparing TMC435 doses of 75 and 150 mg QD (the doses chosen for the ongoing Phase IIb trial, PILLAR), this 2-fold increase in dose is likely to equate to a disproportionate increase in TMC435 exposure, with the median AUC and C_{max} values expected to increase approximately 4-fold.

Figure 3. Change from baseline to Day 28 in (A) total, (B) direct and (C) indirect bilirubin; (D) alkaline phosphotase (ALP); (E) aspartate aminotransferase (AST) and (F) alanine aminotransferase (ALT) levels as a function of TMC435 exposure (defined as AUC_{24h}).

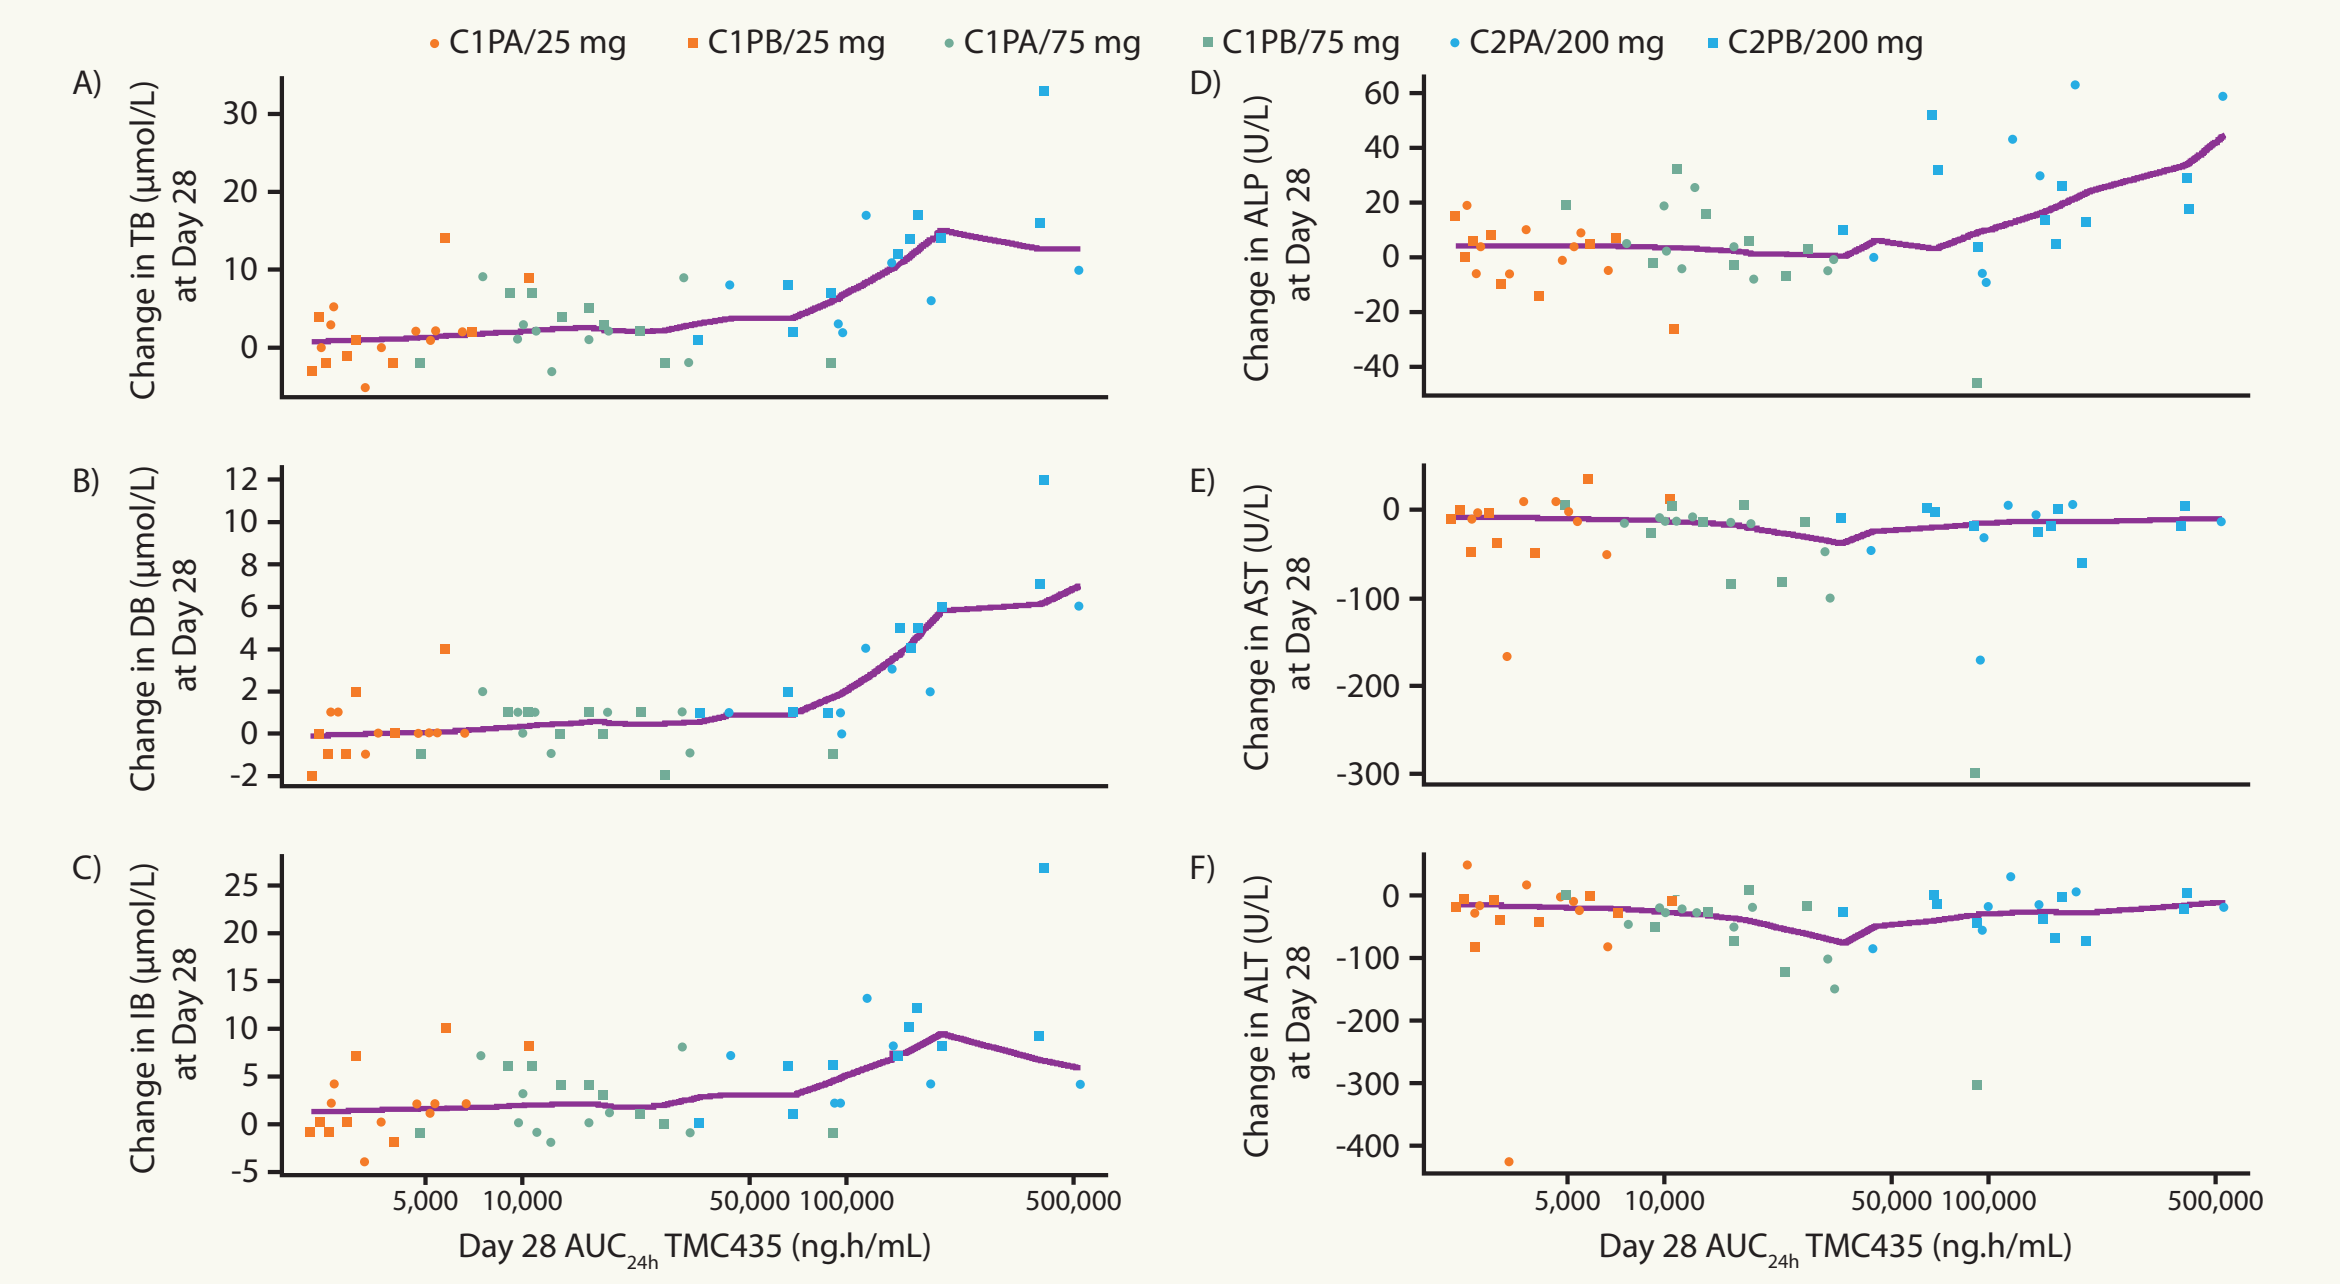

C1PA/25 mg
C1PB/25 mg
C1PA/75 mg
C1PB/75 mg
C2PA/200 mg
C2PB/200 mg
A)
Change in TB (µmol/L) at Day 28
B)
Change in DB (µmol/L) at Day 28
C)
Change in IB (µmol/L) at Day 28
D)
Change in ALP (U/L) at Day 28
E)
Change in AST (U/L) at Day 28
F)
Change in ALT (U/L) at Day 28
5,000 10,000
50,000 100,000
500,000
Day 28 AUC24h TMC435 (ng.h/mL)

Lines represent the LOESS spline fitted to the data
AUC_{24h}, area under the concentration-time curve from time of administration up to 24 hours; C, cohort; DB, direct bilirubin; IB, indirect bilirubin; PA, panel A; PB, panel B; TB, total bilirubin

Conclusions

- **In treatment-naive HCV-infected patients, following 28 days of treatment with TMC435 in combination with P/R, a dose-proportional increase in TMC435 exposure was observed from 25 to 75 mg QD, with a more than dose-proportional increase in TMC435 exposure observed from 75 to 200 mg QD.**
- **There was no clear PK-PD relationship between TMC435 exposure and antiviral activity with TMC435 doses of 75 mg QD or higher.**
- **A trend towards mild increases in serum bilirubin and ALP was observed with higher TMC435 exposure, with increases mainly observed at the 200 mg dose. However, no clear PK-PD relationship was observed for other liver parameters.**
- **The PK-PD findings reported here support the TMC435 75 and 150 mg QD doses selected for further evaluation in treatment-naive patients as part of the ongoing Phase IIb trial, PILLAR.**

References

1. Lin TI et al. Antimicrob Agents Chemother 2009; 53: 1377–1385.
2. Reesink HW et al. Gastroenterology 2010; 138: 913–921.
3. Reesink HW et al. Poster presented at the 60th American Association for the Study of Liver Diseases (AASLD) meeting, Boston, MA, USA, 30 October–3 November, 2009.
4. Marcellin P et al. Poster presented at the 44th Annual Meeting of European Association for the Study of the Liver (EASL), Copenhagen, Denmark, 22–26 April, 2009.
5. Manns M et al. Presented at the 44th Annual Meeting of European Association for the Study of the Liver (EASL), Copenhagen, Denmark, 22–26 April, 2009.

Poster presented at the 45th Annual Meeting of the European Association for the Study of the Liver (EASL), Vienna, Austria, 14–18 April, 2010